Novel Amyloid Beta Monoclonal Antibodies With Superior Binding Properties: Potential for More Convenient Dosing and Greater Patient Access in Alzheimer’s Disease

Michael Skov, Robin Barbour, Philip Dolan, Abderrahman Elmaarouf, Erich Goldbach, Michael Holden, Lauri Li, Tarlochan Nijjar, Heather Prill, Joshua Salmans, Stephen Tam, Kimberly Thomas, Clara Tourino, Frédérique Bard, Gene G. Kinney, **Wagner Zago**

Prothena Biosciences Inc, South San Francisco, CA, USA

INTRODUCTION

- Monoclonal antibodies (mAbs) targeting the N-terminus of amyloid beta (Aβ) have been demonstrated clinically to reduce amyloid plaque burden[1]
- One such antibody currently under FDA priority review, aducanumab, showed that significant reduction in plaque burden was associated with reduction in clinical decline in Alzheimer’s disease (AD)[2]
- Preclinical studies have also indicated that N-terminal mAbs elicit antibody-dependent microglial-mediated Aβ-plaque clearance and neutralization of soluble toxic Aβ aggregates in vitro and/or in vivo[3,4]
 - It is hypothesized that administration of N-terminal Aβ targeting mAbs slows disease progression via clearance of Aβ plaques and neutralization of soluble Aβ aggregates in patients with AD

AIM

- To develop and characterize novel humanized N-terminal Aβ-targeting antibodies with greater binding strength (affinity and avidity) for pathologic fibrillar Aβ than has been reported with current experimental therapies and with high affinity for soluble toxic forms
- Antibodies with such characteristics may be efficacious therapies for AD and could enable more convenient dosing strategies, which will enhance patient access

METHODS

Generation of Antibodies

- The full amino acid sequence of aducanumab was selected based on publicly available sequence information, and was used to generate the humanized antibody
 - Aducanumab and Prothena (PRO) mAbs (h2726, h2731, h2831, h2931) were expressed in Chinese hamster ovary (CHO) cells, purified by protein A affinity chromatography, and characterized by capillary electrophoresis and size exclusion chromatography

Surface Plasmon Resonance (SPR) Measurements

- Binding kinetics were performed with a Biacore T200 (GE Healthcare, Princeton, NJ)
 - Aβ42 hexafluoroisopropanol (HFIP) films (Bachem, Torrance, CA) were resuspended in DMSO to 5 mM and further diluted to 100 μM with 10 mM HCl
 - Samples were incubated at 37°C for 24 hours, then centrifuged to separate soluble and fibrillar species
 - The pellet was resuspended in 1x Dulbecco’s phosphate-buffered saline (PBS) and sonicated before use
 - Fibrils were immobilized on sensor chip CM5 (GE Healthcare Life Sciences, Princeton, NJ)
 - Serially diluted antibodies were injected at 30 μL/min in running buffer (HEPES buffered saline + 0.05% P-20, 1 mg/mL bovine serum albumin [BSA]) for 300 seconds association time and 1200 seconds dissociation time
 - Analysis was performed using a global 1:1 fit with Biacore Insight Evaluation software (v2.0)

Binding of Aβ Fibrils by Enzyme-Linked Immunosorbent Assay (ELISA)

- Aβ42 fibrils were prepared as in SPR assay
 - Aβ42 fibrils 2.5 μg/mL were coated overnight at room temperature (RT)
 - Plates were blocked with 1% BSA/PBS for 1 hour
 - Antibodies were serially diluted in 0.1% BSA/PBS and 0.1% Tween 20 and were incubated for 2 hours at RT in duplicate
 - Plates were washed with TBS/Tween 20, and goat anti-human IgG horseradish peroxidase (HRP; Invitrogen, Carlsbad, CA) was added and incubated 1 hour at RT
 - Plates were washed in TBS/Tween 20, and antibody binding was detected with streptavidin HRP and o-phenylenediamine dihydrochloride (OPD) substrate (Thermo Fisher Scientific, Waltham, MA) following manufacturer’s instructions
 - Plates were read at 490 nm on a SpectroMax (Molecular Devices, San Jose, CA); EC50 estimations represent nonlinear 3 parameter least-squares fit of data using GraphPad Prism software

Immunohistochemistry on AD Tissues

- Frozen human AD brain samples (Banner Sun Health Research Institute, Sun City, AZ) were embedded in Tissue-Tek OCT (Sakura Finetek, Torrance, CA)
 - The staining was performed in an automated stainer (Leica Biosystems, Buffalo Grove, IL)
 - Binding of the biotinylated antibodies was visualized using the avidin-biotin amplification system (ABC Elite Standard, PK-6100; Vector Laboratories, Burlingame, CA)
 - The slides were digitally imaged (NanoZoomer 2.0-HT, Hamamatsu Corporation, Bridgewater, NJ) and analyzed using HALO software (Indica Labs, Albuquerque, NM) to measure percentage of stained tissue, and results were plotted using GraphPad Prism software

Ex Vivo Phagocytosis Assay

- Cryostat sections of APP/PS1 mouse (reMYND) collected on coverslips were incubated with antibodies for 1 hour
 - Primary mouse microglial cells were then seeded, and the cultures were maintained at 37°C for 72 hours
 - Media was carefully aspirated and sections washed with PBS
 - The sections were resuspended in 8M urea, and Aβ1-42 was quantified using a V-PLEX Aβ42 Peptide (4G8) Kit (Meso Scale Discovery, Gaithersburg, MD)

Binding of Soluble Aβ Aggregate to Rat Hippocampal Neurons

- Primary rat hippocampal neurons and soluble Aβ aggregates were prepared as described[4]
 - Soluble Aβ (1 μM) was pre-incubated with antibodies or isotype control for 30 minutes at 37°C in Neurobasal-no phenol red (NB-NPR) (Thermo Fisher Scientific)
 - The antibody/Aβ complexes were added to cells and incubated for 30 minutes at 37°C
 - The neurons were rinsed in NB-NPR, fixed in 4% paraformaldehyde, and immunostained with mouse monoclonal anti-Aβ antibody MABN254 (EMD Millipore, Burlington, MA)
- Binding of Aβ was quantified by high-content quantitative imaging analysis on an Operetta CLS (PerkinElmer, Waltham, MA) instrument using modified neurite outgrowth algorithm with microtubule-associated protein 2 (Abcam; Cambridge, UK), NeuN, and mAb Aβ1-42 (EMD Millipore) primary antibodies followed by Alexa Fluor (Thermo Fisher Scientific) secondary detection antibodies; 40x water objective; 25 optical fields per well in 96-well microplate format

RESULTS

Figure 1. Measure of Direct Binding and Relative Affinity of Antibodies to Fibrillar Aβ42 by ELISA

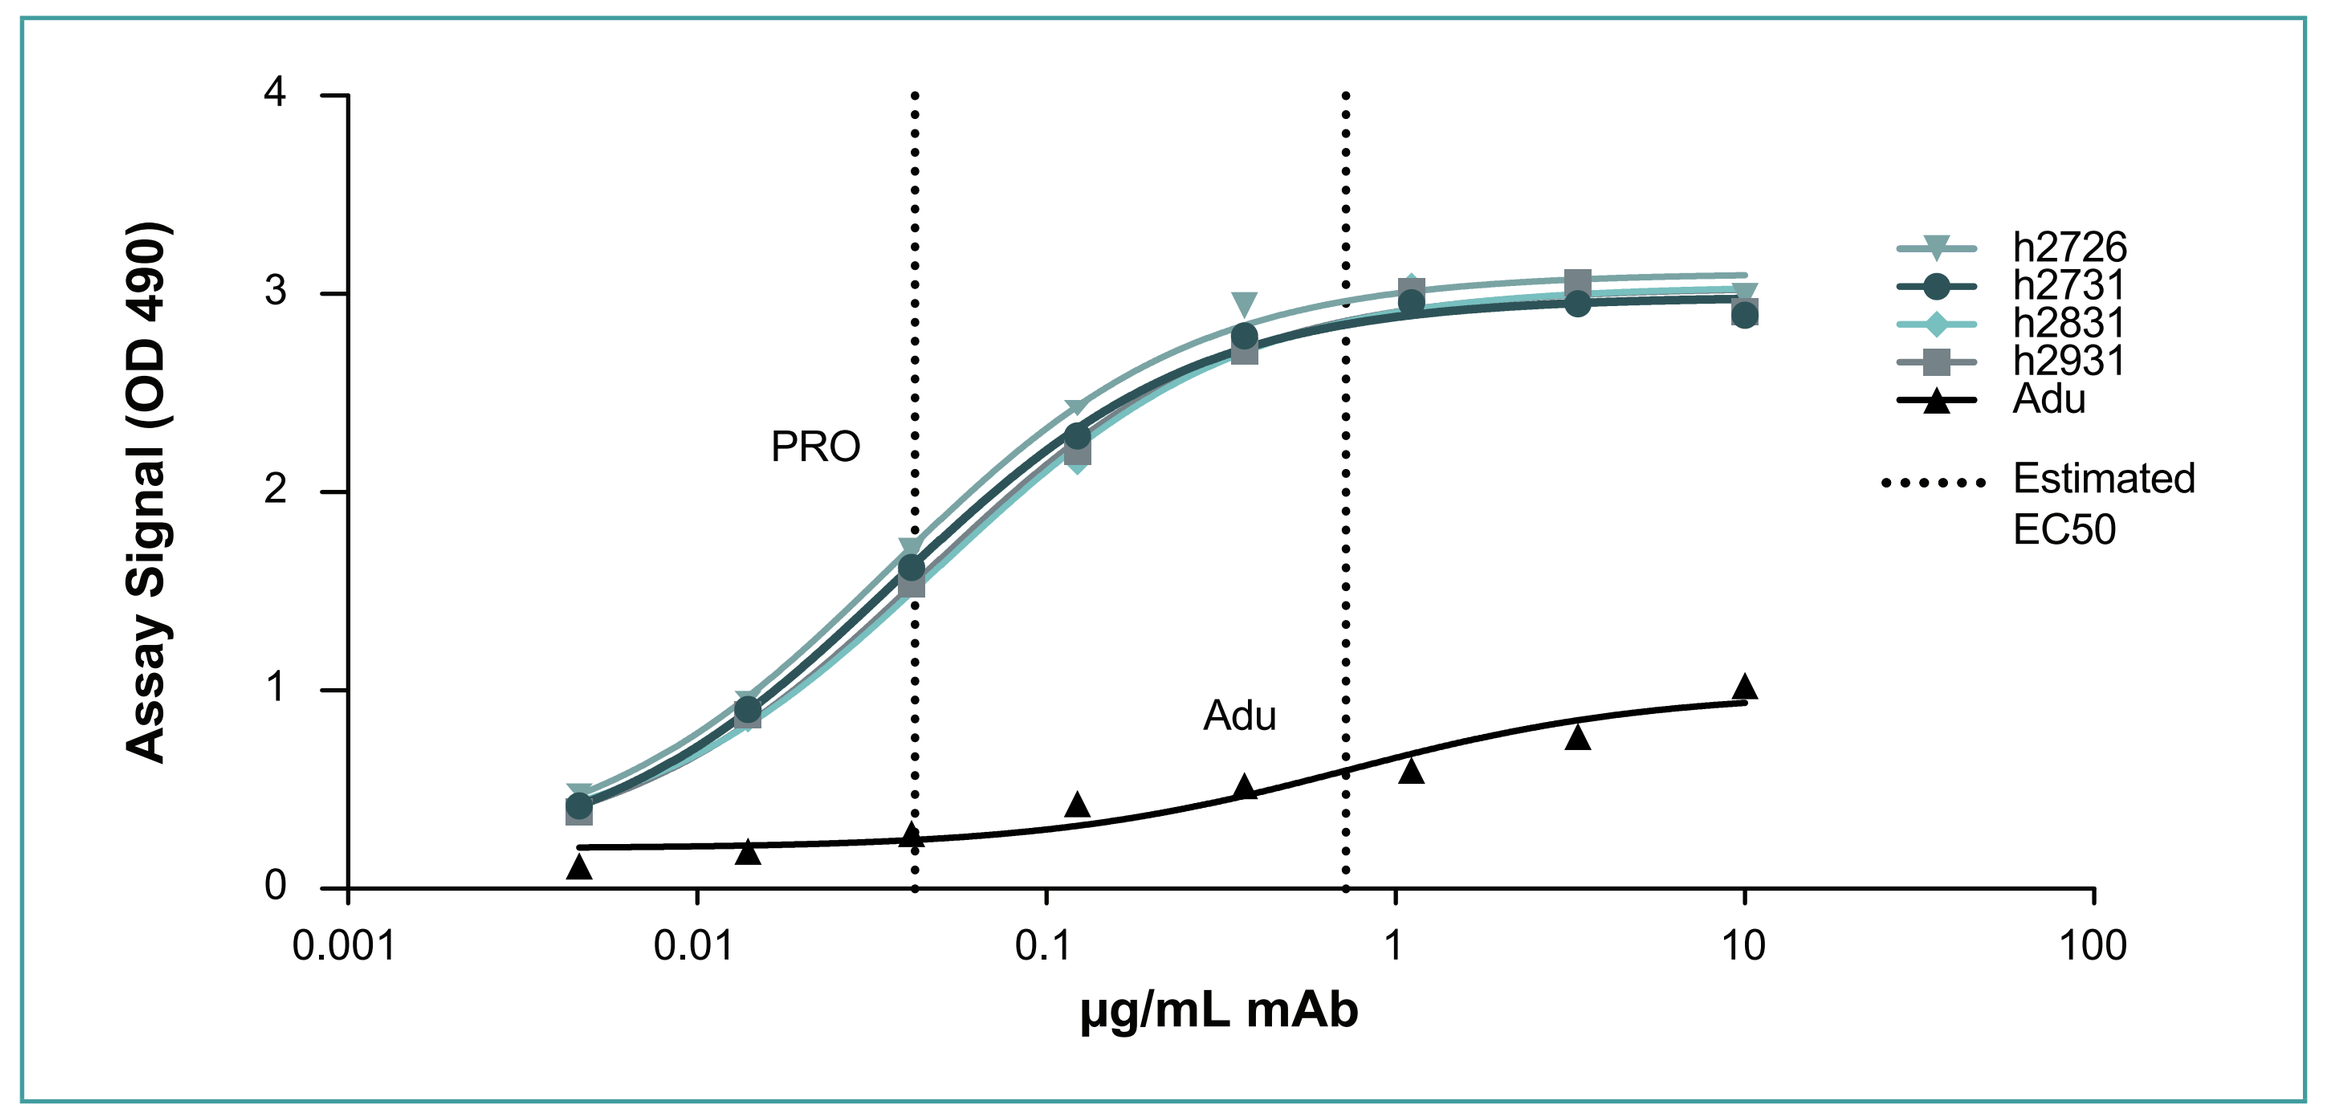

Assay Signal (OD 490)
μg/mL mAb
PRO
Adu
h2726
h2731
h2831
h2931
Adu
Estimated EC50

Aβ42, amyloid beta 42; Adu, aducanumab; EC50, effective concentration 50%; ELISA, enzyme-linked immunosorbent assay; mAb, monoclonal antibody; OD 490, optical density 490 nanometer; PRO, Prothena antibodies.
Curves and resulting EC50 estimations represent nonlinear 3-parameter least-squares fit of data. Individual points are average of duplicate samples (coefficient of variation <8%).

- The direct binding of PRO mAbs and aducanumab to Aβ42 fibrils was assessed by ELISA (**Figure 1**)
 - A 3-fold increase in assay signal (OD 490) and 15- to 20-fold lower estimated EC50 indicated increased overall binding and relative avidity of PRO mAbs to fibrillar Aβ relative to aducanumab

Table 1. Kinetic Binding Parameters of Antibodies to Fibrillar Aβ Determined by SPR

Analyte (mAb)	ka (1/Ms)	kd (1/s)	KD* (M)	KD* (pM)	R_{max}
Adu	2.96e+7	1.70e-2	5.74e-10	574	45.2
h2726	3.93e+5	2.12e-5	5.40e-11	54.0	51.0
h2731	3.72e+5	2.62e-5	7.04e-11	70.4	50.7
h2831	2.65e+5	2.94e-5	1.11e-10	111	50.2
h2931	3.35e+5	2.05e-5	6.12e-11	61.2	50.0

Aβ, amyloid beta; Ka, association rate constant; kd, dissociation rate constant; KD*, apparent equilibrium dissociation constant; mAb, monoclonal antibody; R_{max}, maximum response; SPR, surface plasmon resonance.

Figure 2. SPR Sensorgram Kinetic Binding Profiles of Antibodies to Fibrillar Aβ at 100 nM Concentration

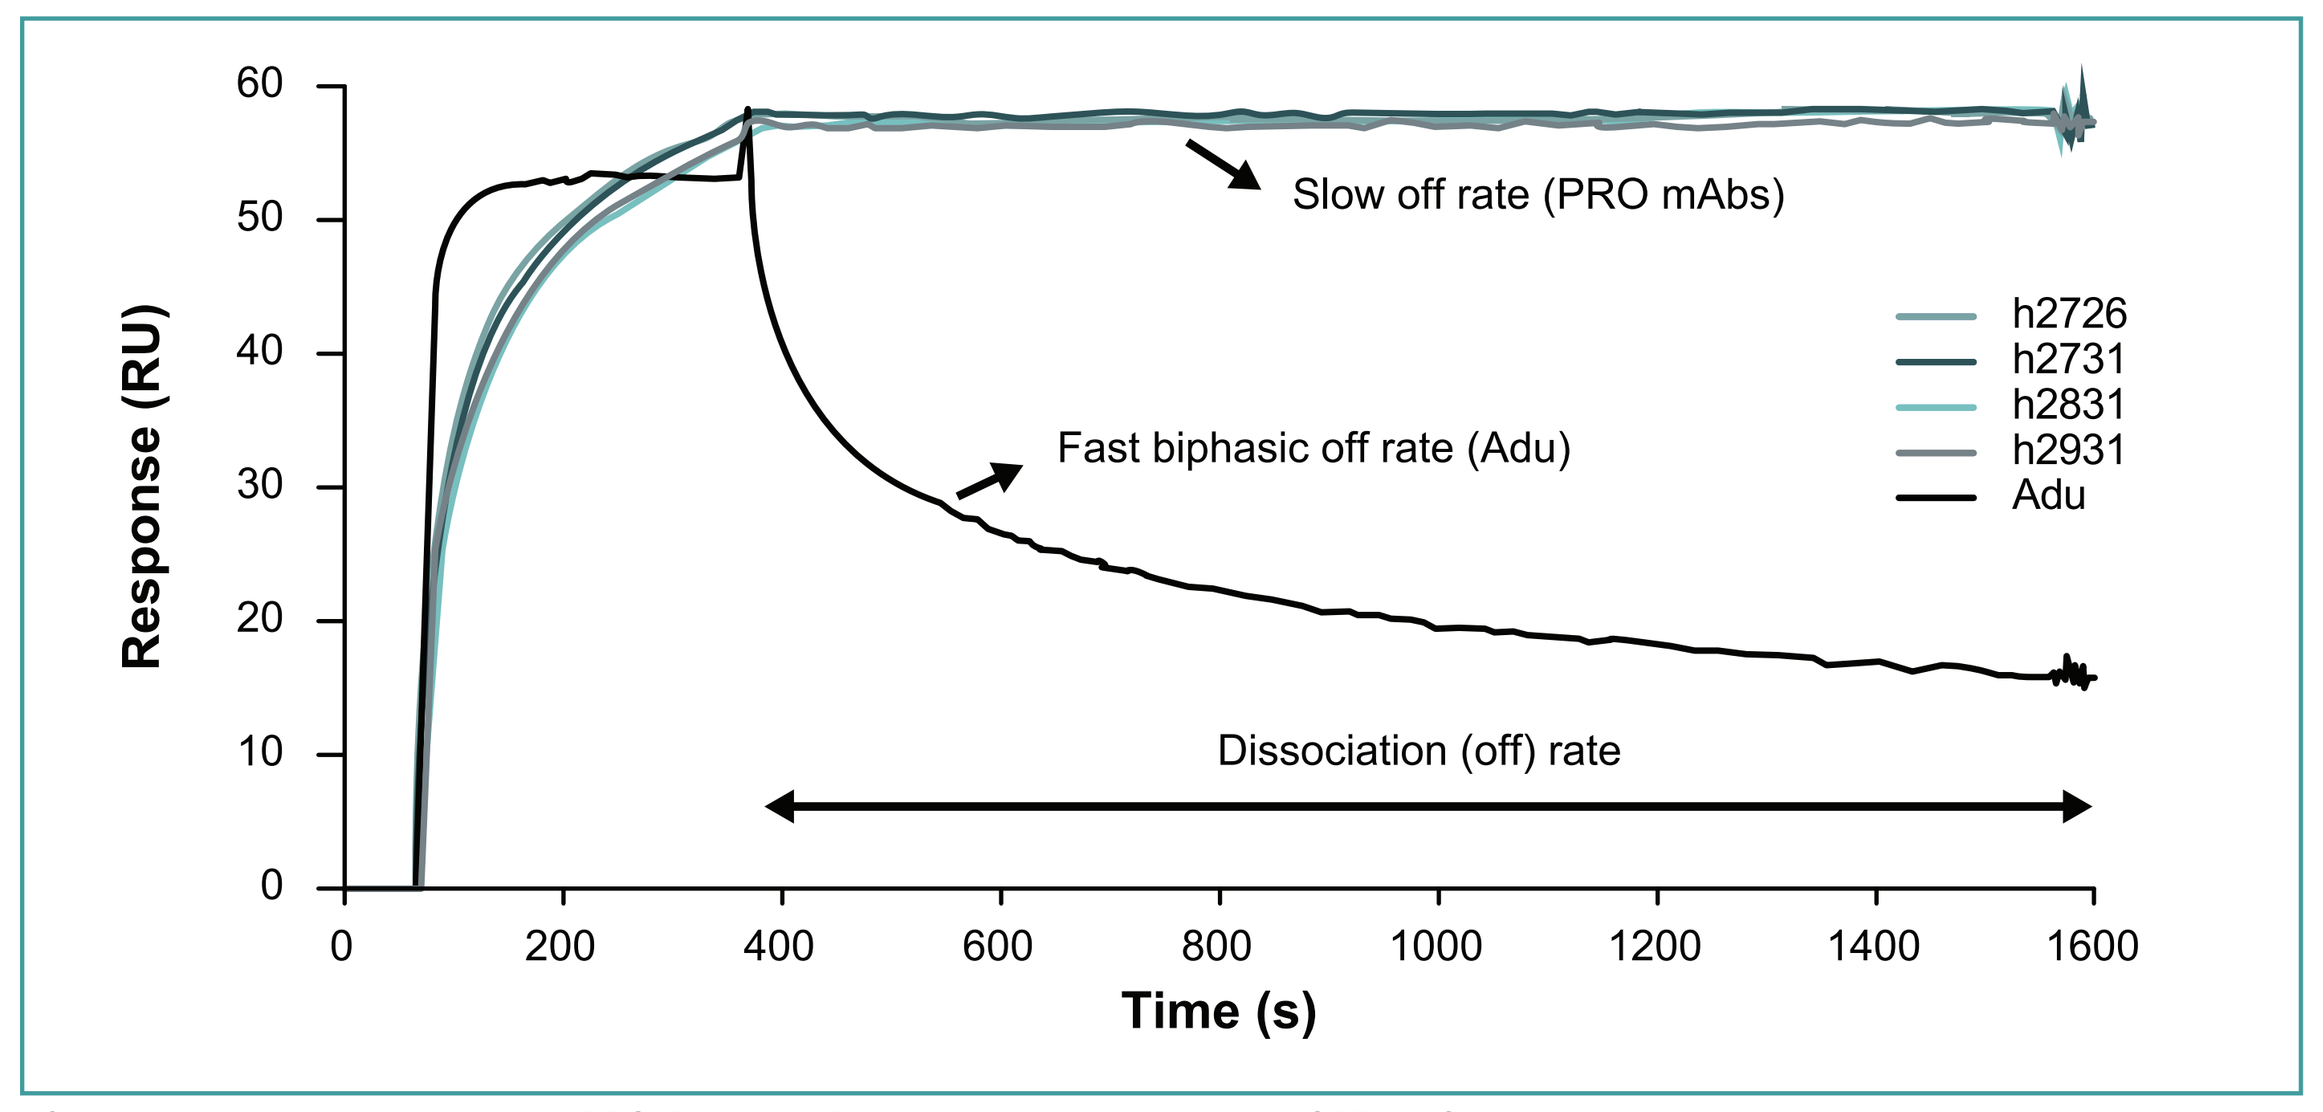

Response (RU)
Time (s)
Slow off rate (PRO mAbs)
Fast biphasic off rate (Adu)
Dissociation (off) rate
h2726
h2731
h2831
h2931
Adu

Aβ, amyloid beta; Adu, aducanumab; PRO, Prothena; RU, response units; s, seconds; SPR, surface plasmon resonance.

- The enhanced relative avidity of PRO mAbs for fibrillar Aβ observed by ELISA was confirmed by SPR equilibrium binding kinetics (**Table 1**), which indicated a 5- to 11-fold greater avidity (apparent KD) than aducanumab
 - This is explained by the different kinetic binding profiles observed in the SPR sensorgram (**Figure 2**)
 - Although aducanumab binds Aβ fibrils at a faster association rate (ka), the much slower dissociation rate (kd) of PRO mAbs resulted in greater measured avidity (ie, lower KD*) than aducanumab

Figure 3. Antibody Dose Response of Aβ Plaque Area Binding Measured as % Positive Tissue by Immunohistochemical Staining in AD Brain

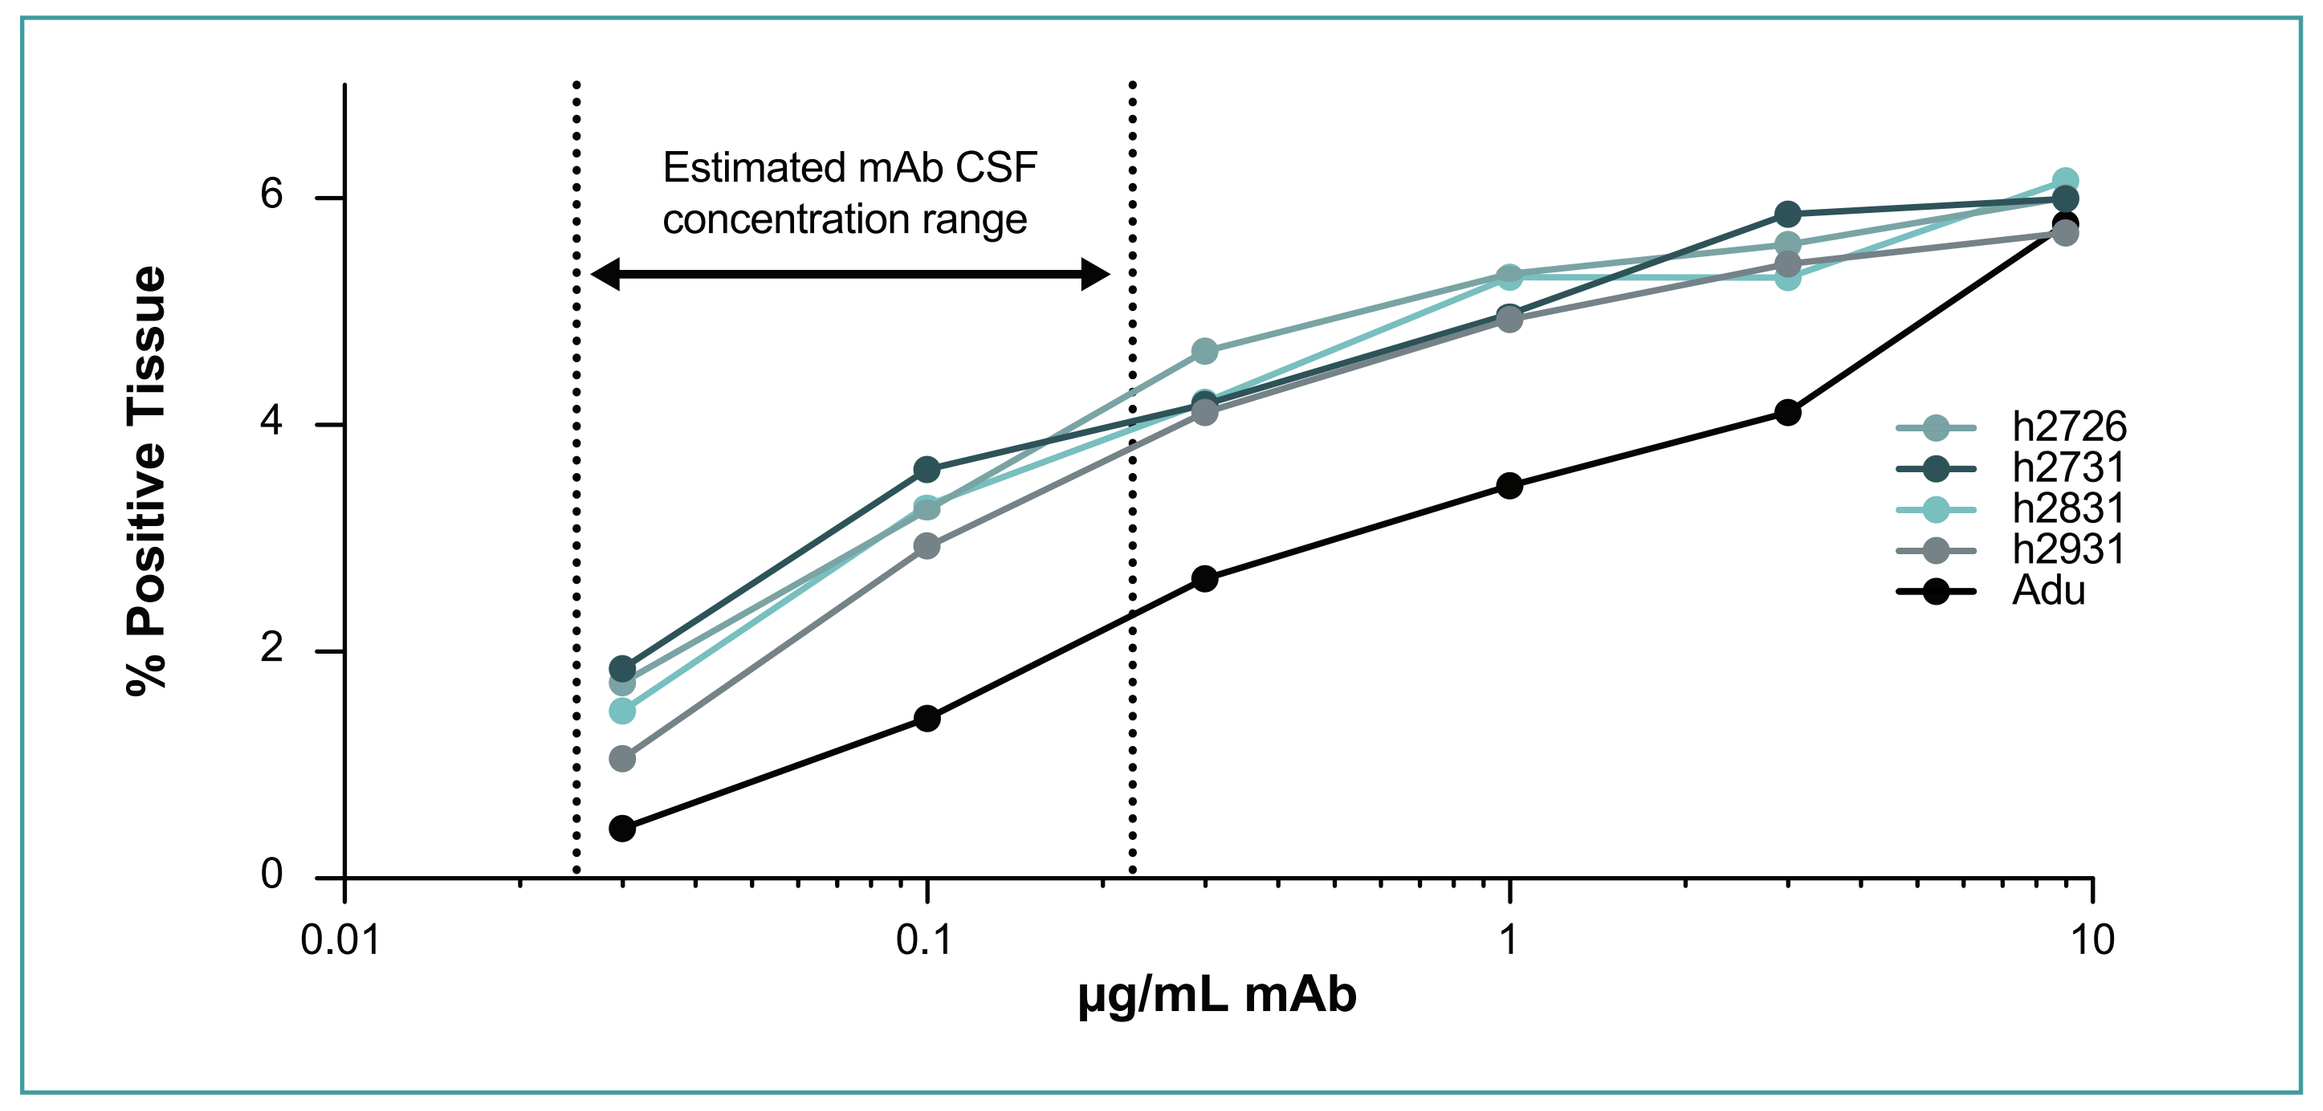

% Positive Tissue
μg/mL mAb
Estimated mAb CSF concentration range
h2726
h2731
h2831
h2931
Adu

Aβ, amyloid beta; AD, Alzheimer’s disease; Adu, aducanumab; CSF, cerebrospinal fluid; mAb, monoclonal antibody.
mAb CSF range estimated as 0.1% of steady-state plasma minimum and maximum concentrations modeled from aducanumab pharmacokinetics at the 10 mg/kg therapeutic dose level.

Figure 4. Representative Aβ Plaque Staining in Individual AD Brain Sections at 0.3 μg/mL of Antibody

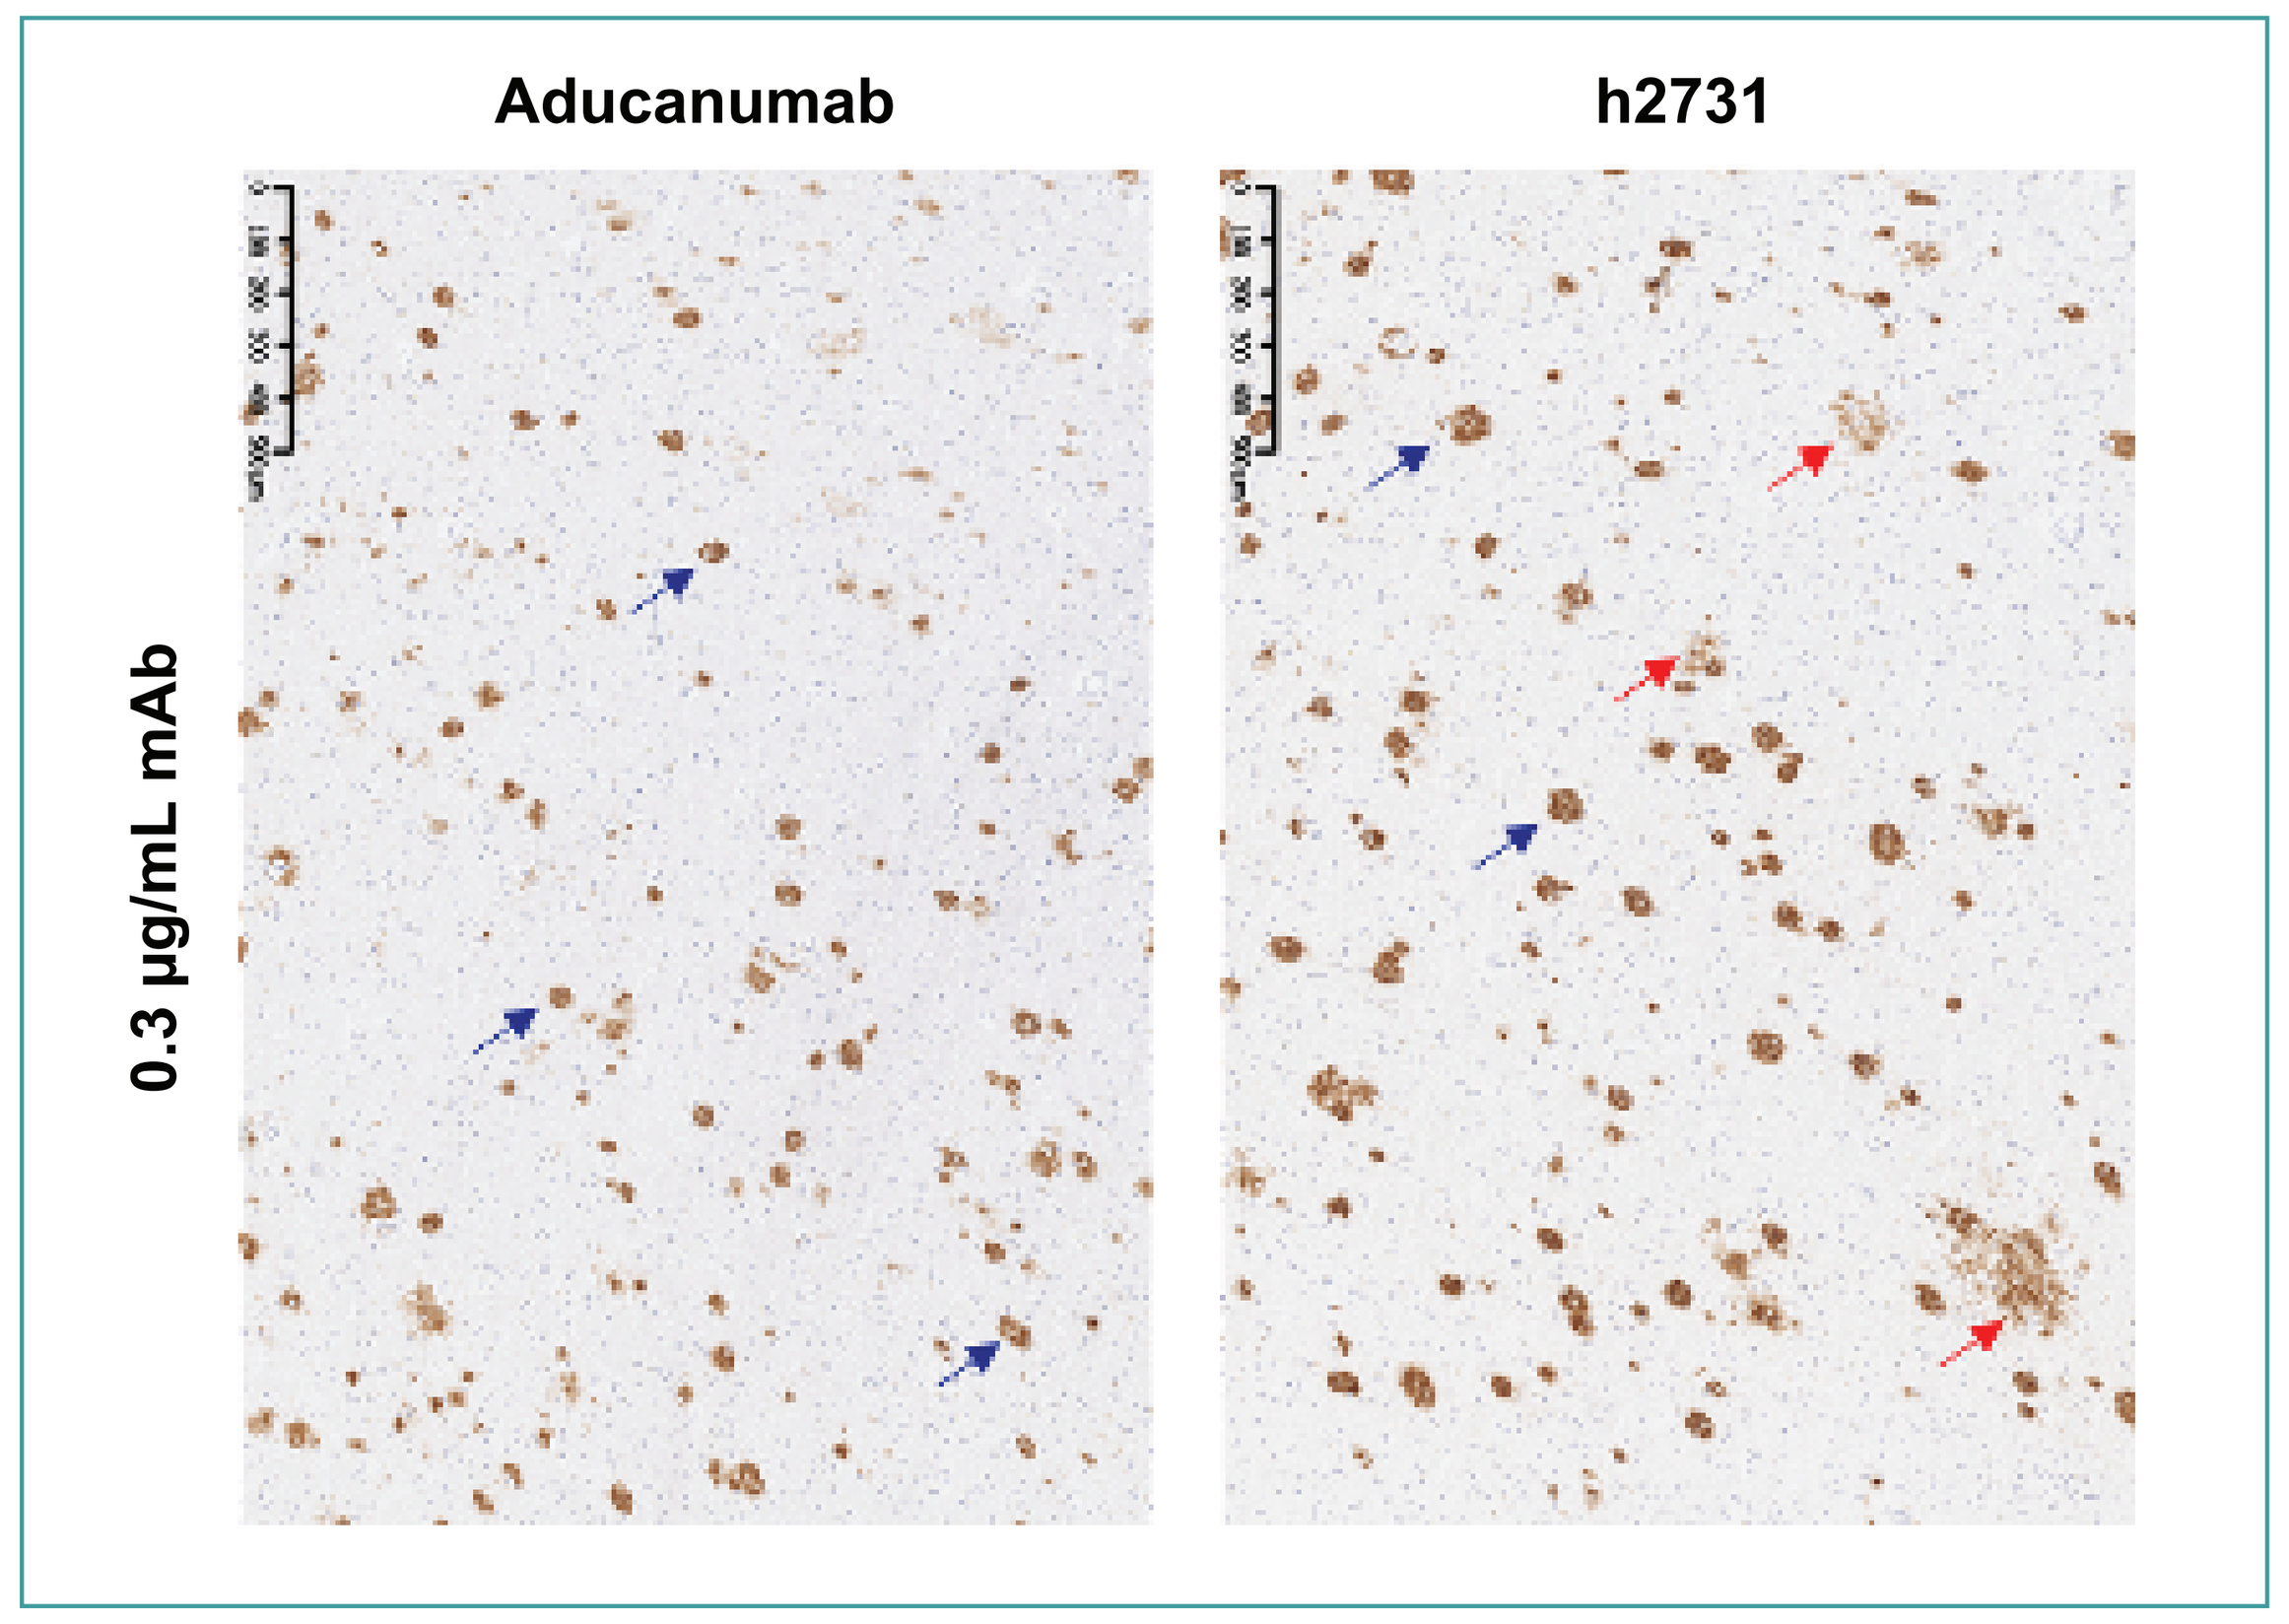

Aducanumab
h2731
0.3 μg/mL mAb

Aβ, amyloid beta; AD, Alzheimer’s disease; mAb, monoclonal antibody.
500 μm scale bar; Blue arrow = compact plaque; red arrow = diffuse plaque.

- Immunohistochemistry concentration-response assessments with PRO mAbs on frozen human AD brain sections (**Figure 3**) showed greater plaque area binding (percentage positive tissue stained) than aducanumab, notably at lower antibody concentrations that are estimated to be clinically relevant exposures in cerebrospinal fluid with 10 mg/kg aducanumab IV. Similar plaque area staining was observed at the highest concentration tested, suggesting saturation of binding at this level
- Immunostaining of AD brain sections showed reactivity on structures typical of AD amyloid plaque pathology (**Figure 4**); however, PRO mAbs appeared to show more intense staining with distribution of binding to both compact (blue arrows) and diffuse (red arrows) plaque, while staining with aducanumab was restricted to mainly compact amyloid plaques, resulting in lower quantification of overall plaque area staining

Figure 5. Antibody-Mediated Aβ42 Clearance by Phagocytosis in APP/PS1 Tg Mouse Brain Measured by MSD Assay

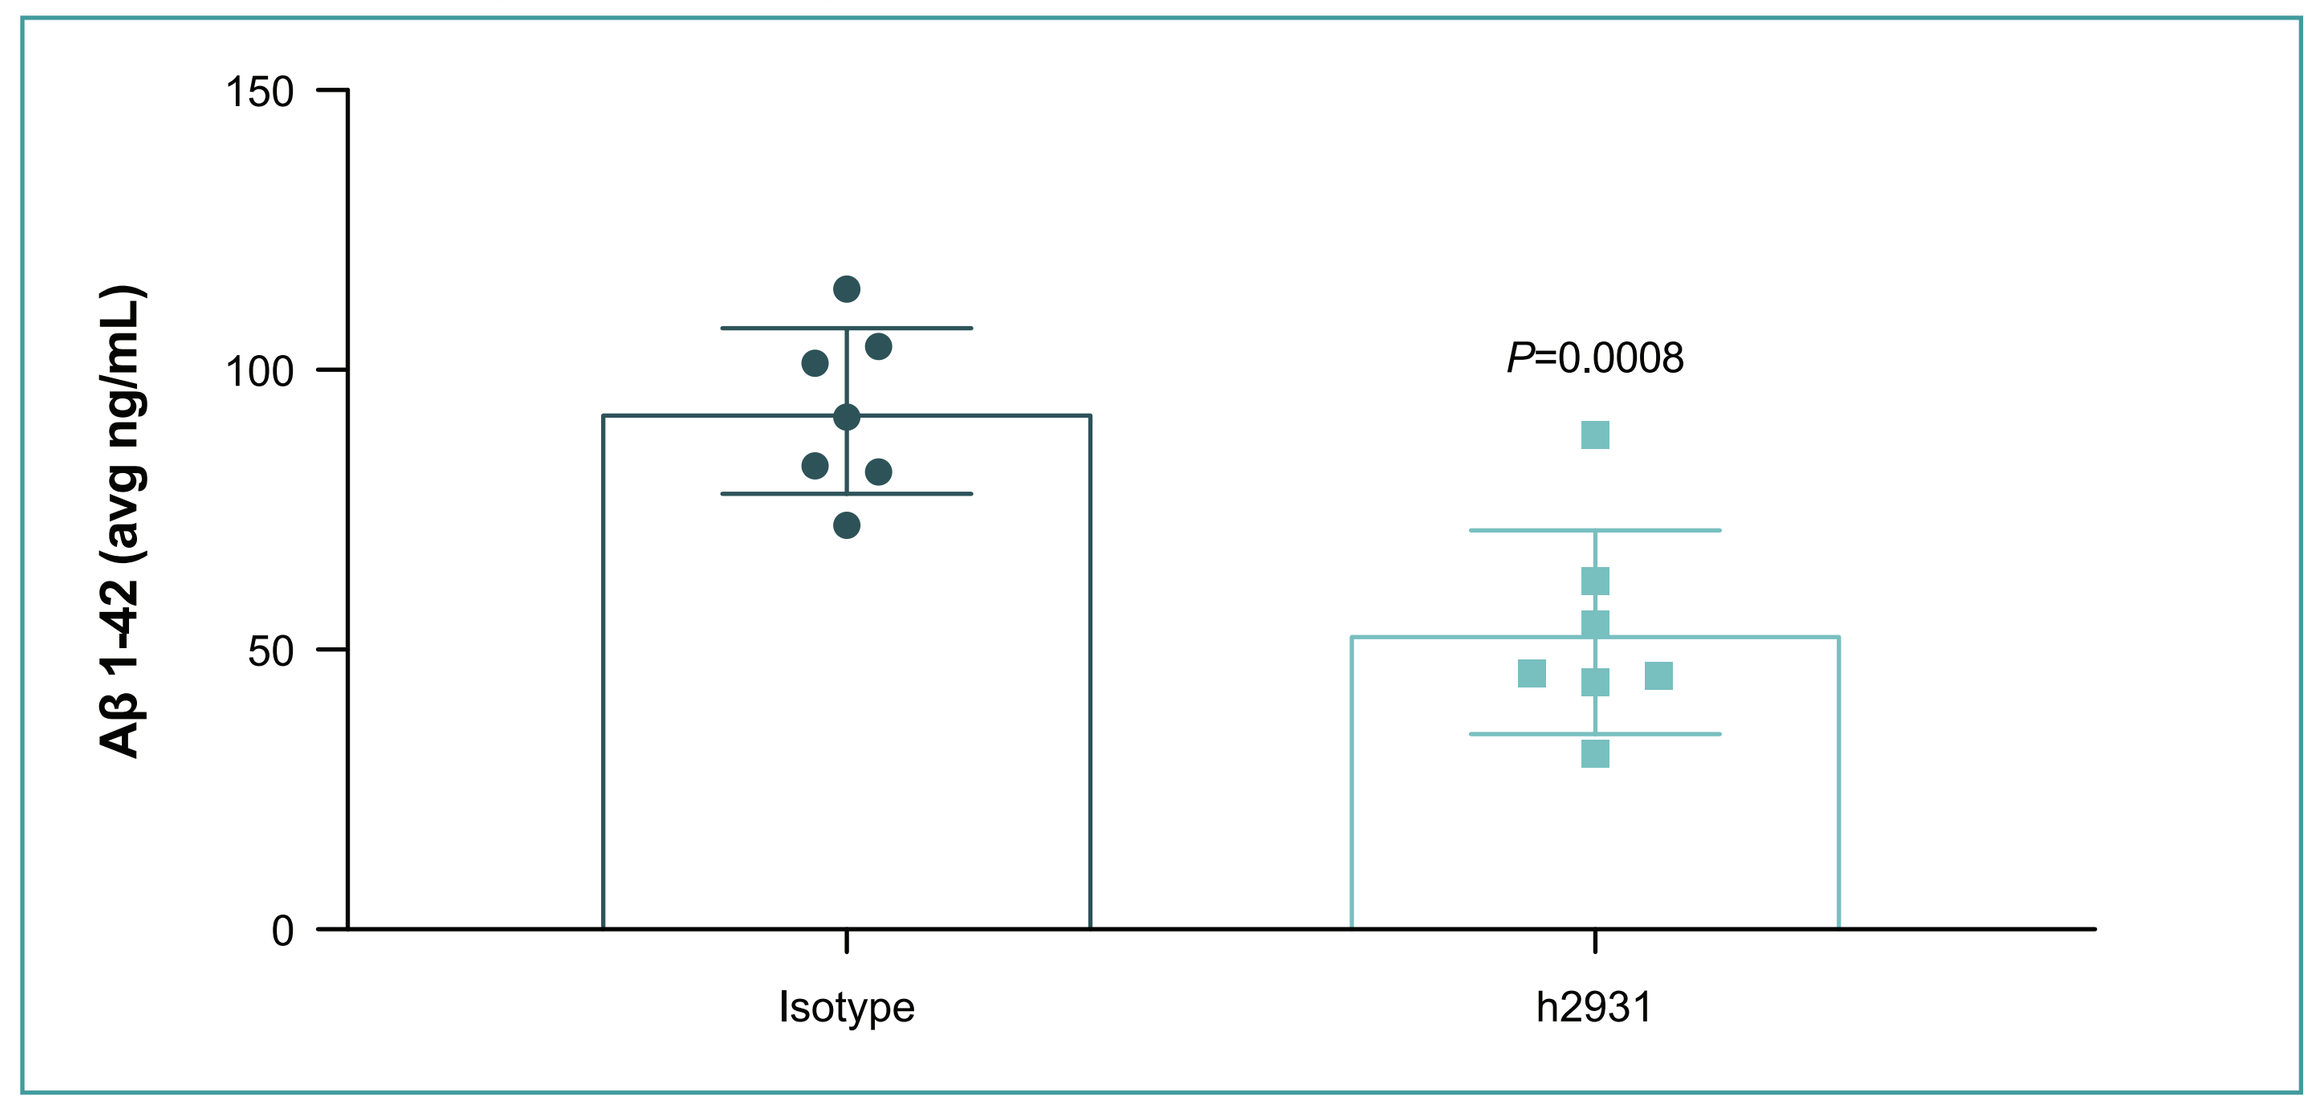

Aβ 1-42 (avg ng/mL)
P=0.0008
Isotype
h2931

Aβ42, amyloid beta 42; MSD, Meso Scale Discovery; SD, standard deviation; Tg, transgenic.
Individual data points shown for each mouse brain tissue section (n=7) represent the average ng/mL Aβ1-42 from 3 experimental replicates.
Bars represent the group mean and SD; *P*=0.0008, unpaired *t* test.

- Using a Meso Scale Discovery assay to quantify brain Aβ42, PRO mAb (h2931) resulted in a significant reduction in amyloid burden mediated by phagocytosis, versus an isotype control in an ex vivo assay using primary mouse microglial cells and frozen brain sections from an APP/PS1 transgenic mouse (**Figure 5**)

Figure 6. Quantification of Binding of Soluble Aβ to Rat Hippocampal Neurons in the Presence of Antibody

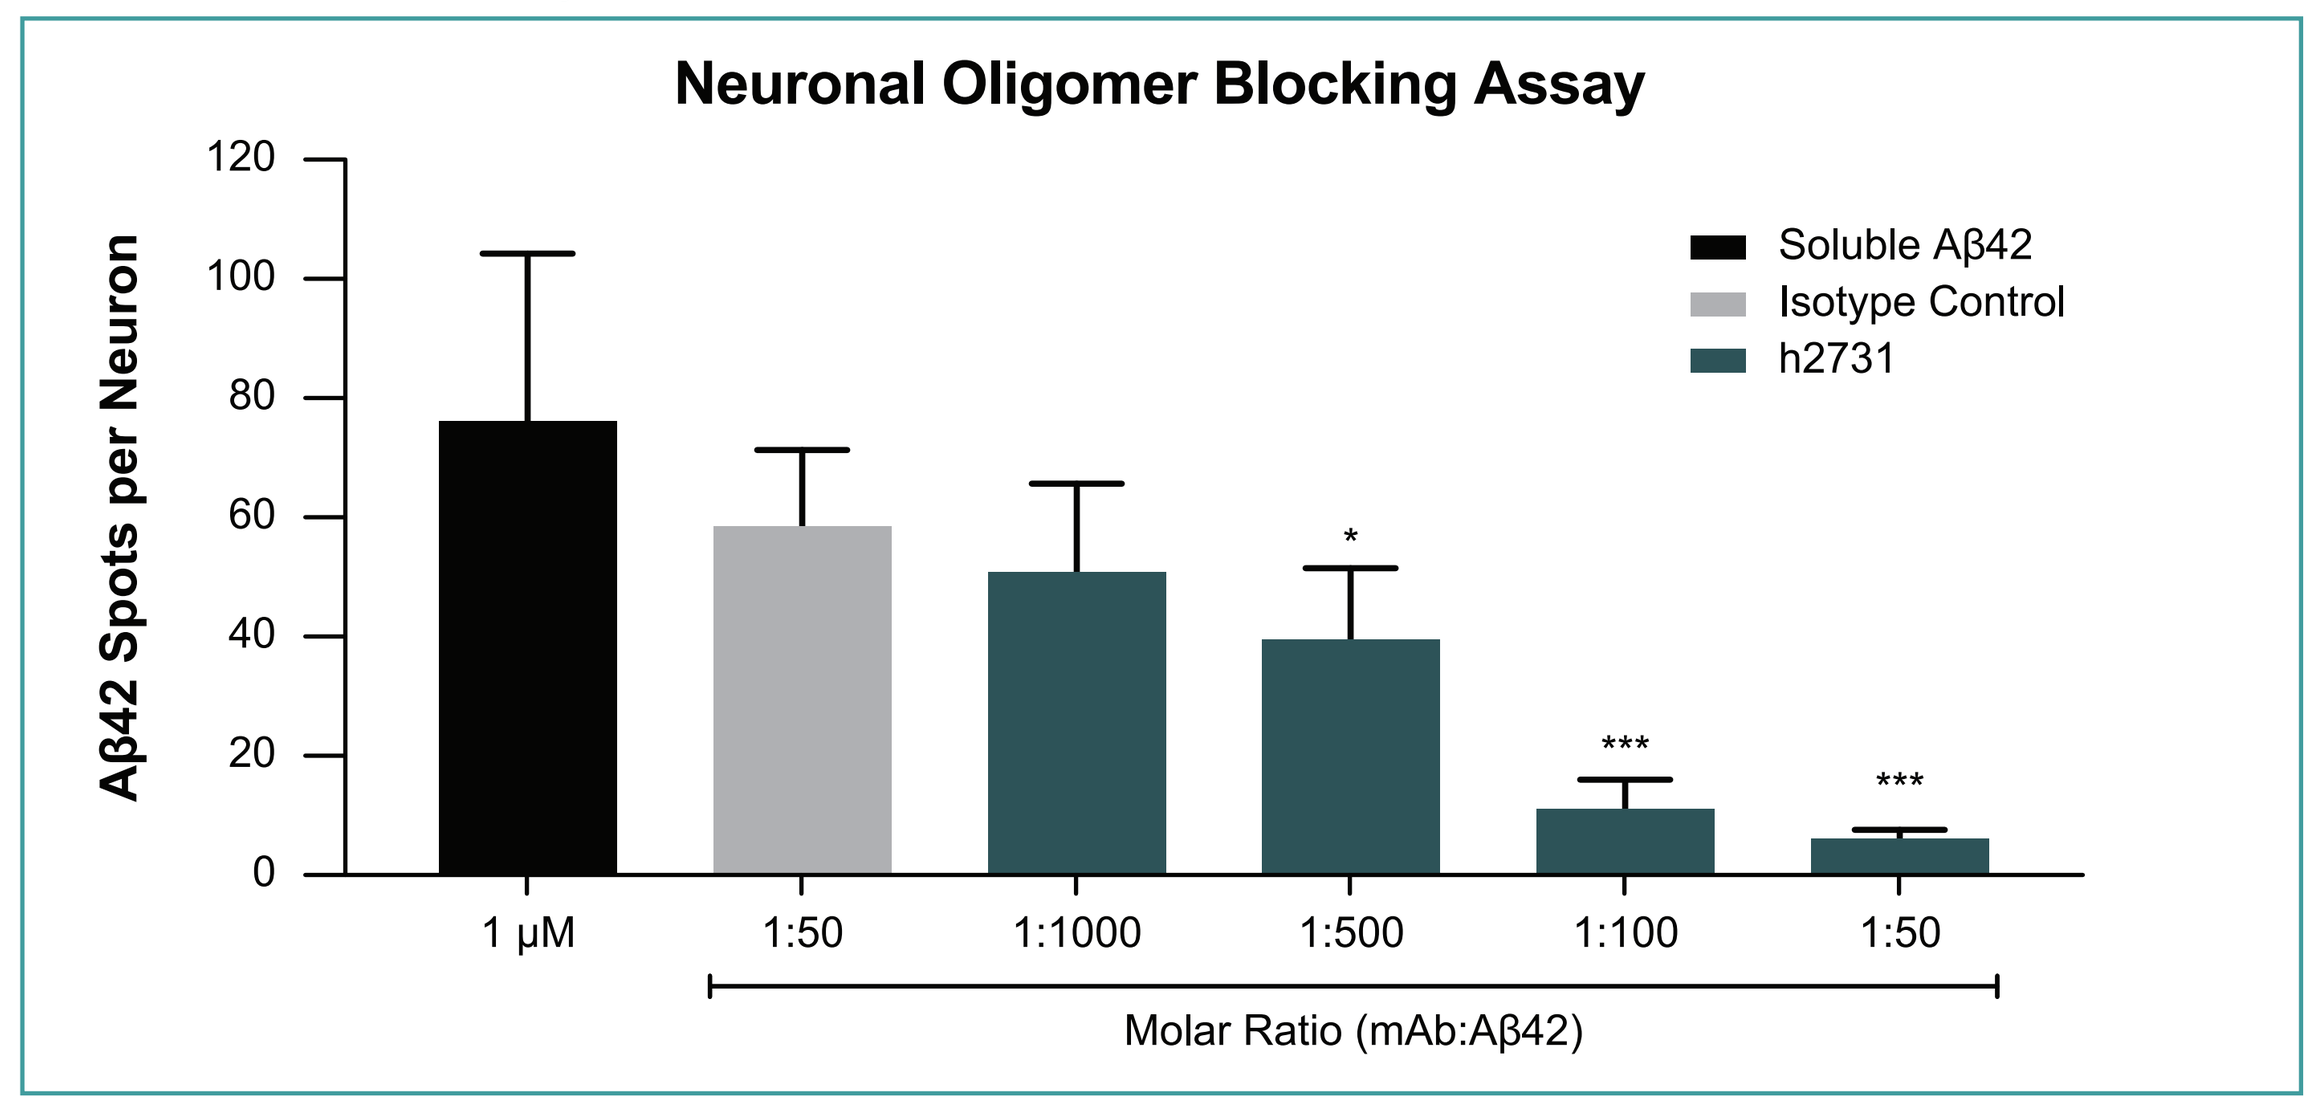

Neuronal Oligomer Blocking Assay
Aβ42 Spots per Neuron
Soluble Aβ42
Isotype Control
h2731
1 μM
1:50
1:1000
1:500
1:100
1:50
Molar Ratio (mAb:Aβ42)

Aβ, amyloid beta; mAb, monoclonal antibody.
Aβ neuritic spots normalized to total neuron number, and results shown as mean ± SD, n=3 wells per condition (150+ neurons each);
*P<0.05, ***P<0.001, one-way ANOVA followed by Dunnett’s multiple comparisons test of individual group means vs mean of Aβ control. Under certain conditions of the assay, aducanumab induced formation of insoluble aggregates that prevented proper quantification of binding to neurons.

- PRO mAb (h2731) effectively blocked the binding of soluble Aβ aggregates to rat hippocampal synapses in a concentration-dependent manner (**Figure 6**)
 - A significant effect of h2731 was observed at a molar mAb:Aβ42 ratio as low as 1:500 (P<0.05) and reached >90% blockade of binding at a 1:50 molar ratio (P<0.001) relative to Aβ42 alone (no mAb pre-incubation)

CONCLUSIONS

- The enhanced Aβ binding properties of PRO mAbs demonstrate their potential as therapeutic candidates that warrant further development. Additionally, it is anticipated the properties of these antibodies will enable evaluation of more convenient dosing strategies in the clinic
- These characteristics could lead to improved patient access and other potential therapeutic benefits

REFERENCES

1. Sevigny J, et al. The antibody aducanumab reduces Aβ plaques in Alzheimer’s disease. *Nature*. 2016;537(7618):50-6. **2.** Budd Haeberlein S, et al. EMERGE and ENGAGE topline results: Two phase 3 studies to evaluate aducanumab in patients with early Alzheimer’s disease. Presented at: CTAD 2019; San Diego, CA; December 5, 2019. **3.** Bard F, et al. Peripherally administered antibodies against amyloid beta-peptide enter the central nervous system and reduce pathology in a mouse model of Alzheimer disease. *Nat Med*. 2000;6(8):916-9. **4.** Zago W, et al. Neutralization of soluble, synaptotoxic amyloid β species by antibodies is epitope specific. *J Neurosci*. 2012;32(8):2696-702.

AUTHOR DISCLOSURES

All authors are employees of Prothena Inc.

ACKNOWLEDGMENTS

This study was sponsored by Othair Prothena Ltd, Dublin, Ireland, a member of the Prothena Corporation plc group. Editorial support was provided by Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ and funded by Prothena Biosciences Ltd.

Presented at the 13th edition of Clinical Trials on Alzheimer’s Disease (CtaD2020), November 4–7, 2020, Virtual/Boston, MA