Poster 74181

PRX005, A Novel Anti-MTBR Tau Humanized Monoclonal Antibody: Results From the Single Ascending Dose Portion of a First-in-Human Double-Blind, Placebo-Controlled, Phase 1 Clinical Trial

Ferenc Martényi, MD,[1] Brian Campbell, PhD,[1] Gene G Kinney, PhD,[1] Jun Lee, PhD,[1] Ann D Johnson, MS,[1] Chad Swanson, PhD,[1] Wagner Zago, PhD,[1] Hideki Garren, MD, PhD[1]

[1]Prothena Biosciences, Inc., South San Francisco, CA, USA

BACKGROUND

- Alzheimer's disease (AD) is a progressive neurodegenerative disease that clinically presents as a gradual onset of dementia, beginning with mild cognitive and functional deficits, leading eventually to an inability to carry out everyday tasks.
- Two hallmark proteins that contribute to AD pathology include amyloid-β and tau. Misfolded and abnormally phosphorylated aggregates of tau protein are the principal proteinaceous component in neurofibrillary tangles.[1] The spread of tau pathology throughout the brain follows a predictable spatiotemporal pattern[2] and is associated with progression of cognitive decline in AD[3].
- Evidence indicates that tau pathology may propagate through cell-to-cell transmission, spreading locally and between interconnected regions of the brain. The extent of tau pathology detected by positron emission tomography (PET) correlates with atrophy and cognitive decline.
- The microtubule-binding region (MTBR) is a primary component of tau tangles and has been empirically determined by Prothena as a superior target region for neuroprotection.[4] Several reports have confirmed the importance of the MTBR in tau pathology:
 - A recent study confirmed the presence of MTBR tau in the cerebrospinal fluid (CSF), and correlated MTBR tau with AD clinical progression.[5]
 - The correlation of MTBR peptides with dementia stages in AD is stronger than that for other tau markers measured (N-terminal, mid-domain).[5]
 - MTBR mediates tau aggregation via a nucleation-dependent mechanism.[6,7]
- PRX005 is an investigational, potential best-in-class humanized IgG1 kappa monoclonal tri-epitopic antibody designed to bind with high affinity to the R1, R2, and R3 repeats in the MTBR of tau, and targets both 3R and 4R tau isoforms.

OBJECTIVES

- The data presented here summarize results from the single ascending dose (SAD) portion of the first-in-human Phase 1 clinical trial.
- The primary objective was to characterize the safety, tolerability, and immunogenicity of PRX005 when administered intravenously (IV) as a single dose in healthy adult subjects.
- The secondary objective was to evaluate the plasma pharmacokinetic (PK) and CSF PK profiles of PRX005.

METHODS

- Healthy volunteers received a single IV dose of PRX005 in 3 cohorts (**Figure 1**):
 - Three dose levels over a modified semi-logarithmic scale were assessed in cohorts S1, S2, and S3.
 - Up to 8 subjects in each dose cohort were randomly assigned to PRX005 or placebo in a 3:1 ratio.

METHODS (CONTINUED)

- Safety data and plasma samples were collected during all visits from day 1 to study completion.
- CSF was sampled in S3 only, at baseline and 2 predefined timepoints.
- Safety follow-up was conducted for up to 2 months.
- Key inclusion and exclusion criteria are described in **Table 1**.

Figure 1. Study Design for SAD Cohorts

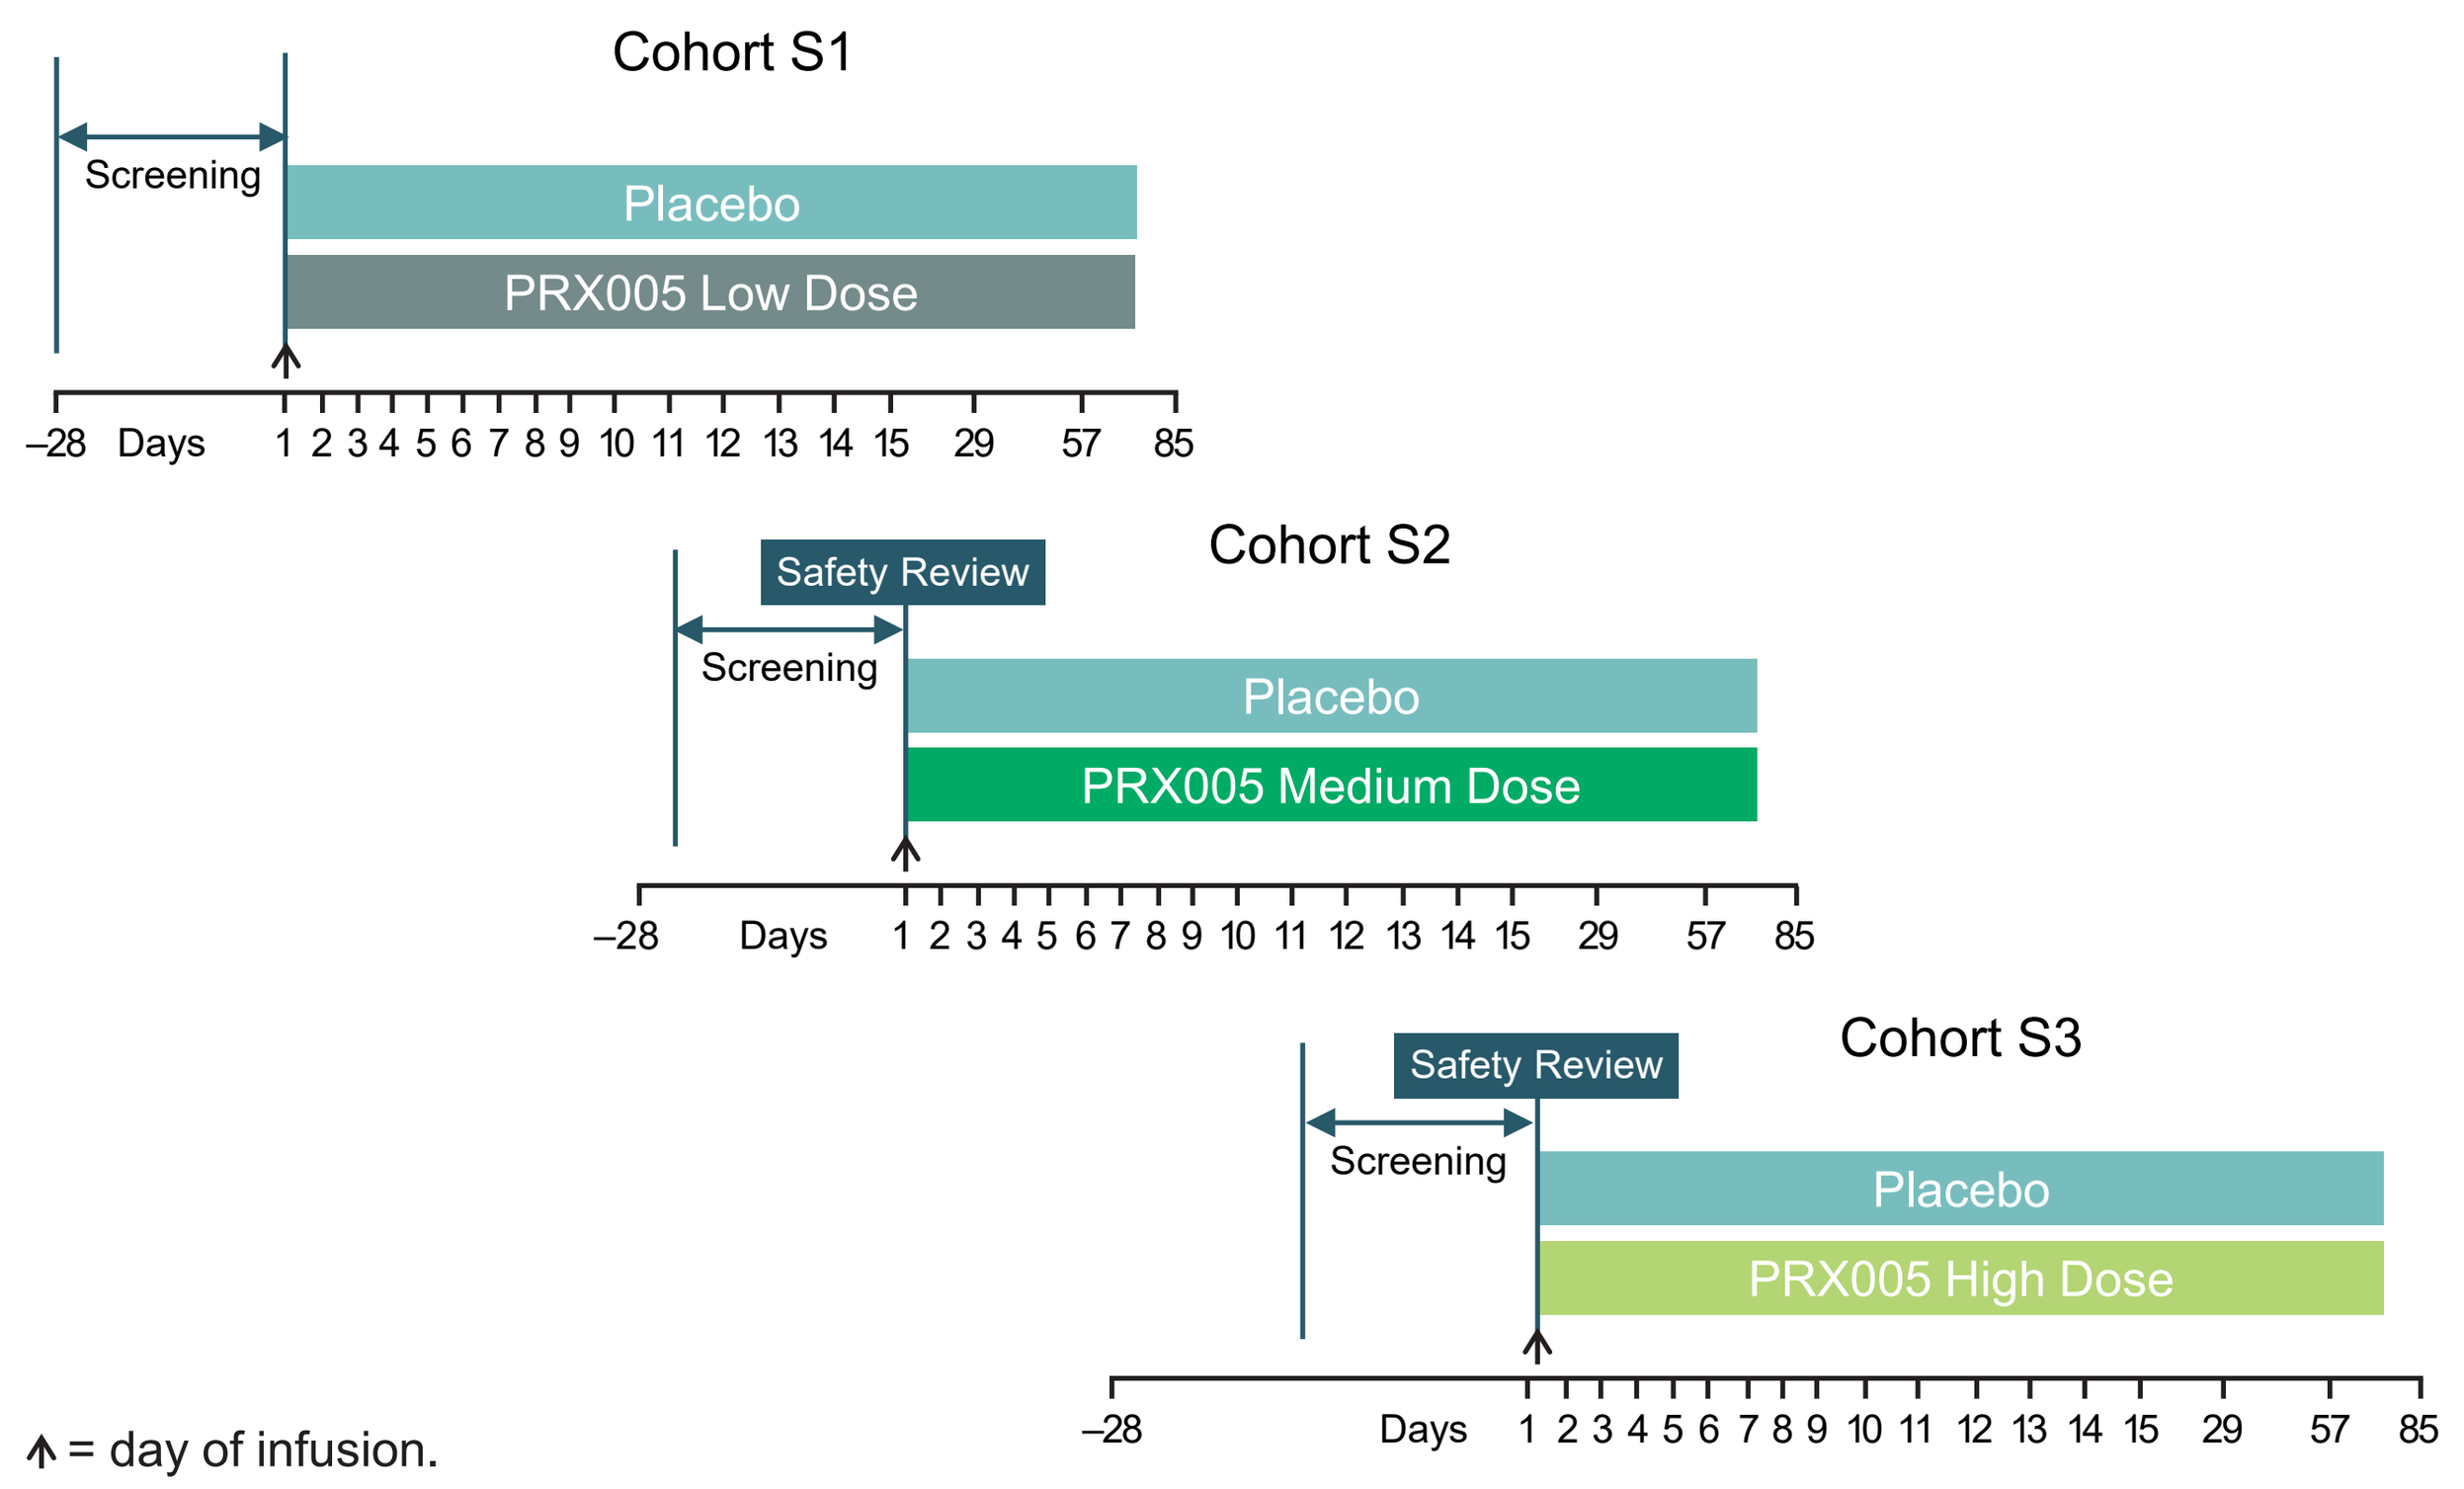
Cohort S1
Screening
Placebo
PRX005 Low Dose
–28 Days 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 29 57 85
Safety Review
Cohort S2
PRX005 Medium Dose
Cohort S3
PRX005 High Dose

↑ = day of infusion.

Table 1. Main Eligibility Criteria

Inclusion Criteria
Healthy subjects between 18 and 65 years of age
Body mass index (BMI) between 18.0 and 32.0 kg/m^2
Exclusion Criteria
General history of any clinically significant gastrointestinal, renal, hepatic, broncho-pulmonary, neurological, psychiatric, cardiovascular, endocrinological, hematological or allergic disease, metabolic disorder, cancer, or cirrhosis
History or presence of systemic autoimmune disorders, or known severe food allergy
History of allergy, hypersensitivity, or serious adverse reaction to monoclonal antibodies or related compounds or allergy to any of the components of the study drug
History of unstable angina, myocardial infarction, advanced chronic heart failure (New York Heart Association Class >2), or clinically significant conduction abnormalities within 1 year before screening
Uncontrolled hypertension or hypotension
History of hereditary short QT syndrome
Fridericia's corrected QT interval (QTcF) >450 msec or the PR interval outside the range of 120 to 220 msec
Malignancy within 5 years

RESULTS

Demographics and Baseline Characteristics

- A total of 25 subjects were randomized and dosed in the study (safety population); 6 assigned to placebo and 19 assigned to PRX005 (low dose, n=7; medium dose, n=6; high dose, n=6).
- One subject in the PRX005 low-dose group withdrew early due to a treatment-emergent adverse event (TEAE) that was not considered related to study drug.
- Demographics and baseline characteristics for the safety population are described in **Table 2.**

Table 2. Demographics and Baseline Characteristics (Safety Population)

	Placebo (n=6)	PRX005 Low Dose (n=7)	PRX005 Medium Dose (n=6)	PRX005 High Dose (n=6)	PRX005 Combined (n=19)
Age at informed consent (years), mean (SD)	51.0 (14.2)	49.6 (15.8)	43.5 (11.6)	45.2 (11.4)	46.3 (12.8)
Sex at birth, n (%)					
Male	6 (100.0)	1 (14.3)	1 (16.7)	4 (66.7)	6 (31.6)
Female	0	6 (85.7)	5 (83.3)	2 (33.3)	13 (68.4)
Ethnicity, n (%)					
Hispanic or Latino	2 (33.3)	3 (42.9)	4 (66.7)	2 (33.3)	9 (47.4)
Not Hispanic or Latino	4 (66.7)	4 (57.1)	2 (33.3)	4 (66.7)	10 (52.6)
Race, n (%)					
Asian	0	0	0	1 (16.7)	1 (5.3)
Black or African American	1 (16.7)	2 (28.6)	0	2 (33.3)	4 (21.1)
White	5 (83.3)	5 (71.4)	6 (100.0)	3 (50.0)	14 (73.7)
Height at screening (cm), mean (SD)	176.5 (6.6)	162.3 (8.5)	163.5 (9.9)	167.7 (11.9)	164.4 (9.8)
Weight at screening (kg), mean (SD)	83.6 (16.9)	69.9 (10.2)	71.4 (12.6)	80.0 (11.3)	73.5 (11.6)
BMI at screening (kg/m^2), mean (SD)	26.7 (4.1)	26.4 (1.9)	26.5 (2.7)	28.4 (2.6)	27.1 (2.5)

SD, standard deviation.

PK and Anti-Drug Antibody (ADA) Results

- Results from SAD cohorts 1–3 indicate PRX005 possesses a PK profile supportive of evaluation of doses in the multiple ascending dose portion of the ongoing clinical trial (**Figure 2; Tables 3 and 4**).
- CSF drug levels after a single dose predict sufficient exposure for saturation of CNS target engagement for >28 days (CSF:plasma ratio=0.2%; **Table 3**).
- No anti-drug antibodies (ADAs) were observed following treatment with PRX005.

RESULTS (CONTINUED)

Figure 2. PRX005 Plasma Concentration–Time Curve Following a Single IV Administration

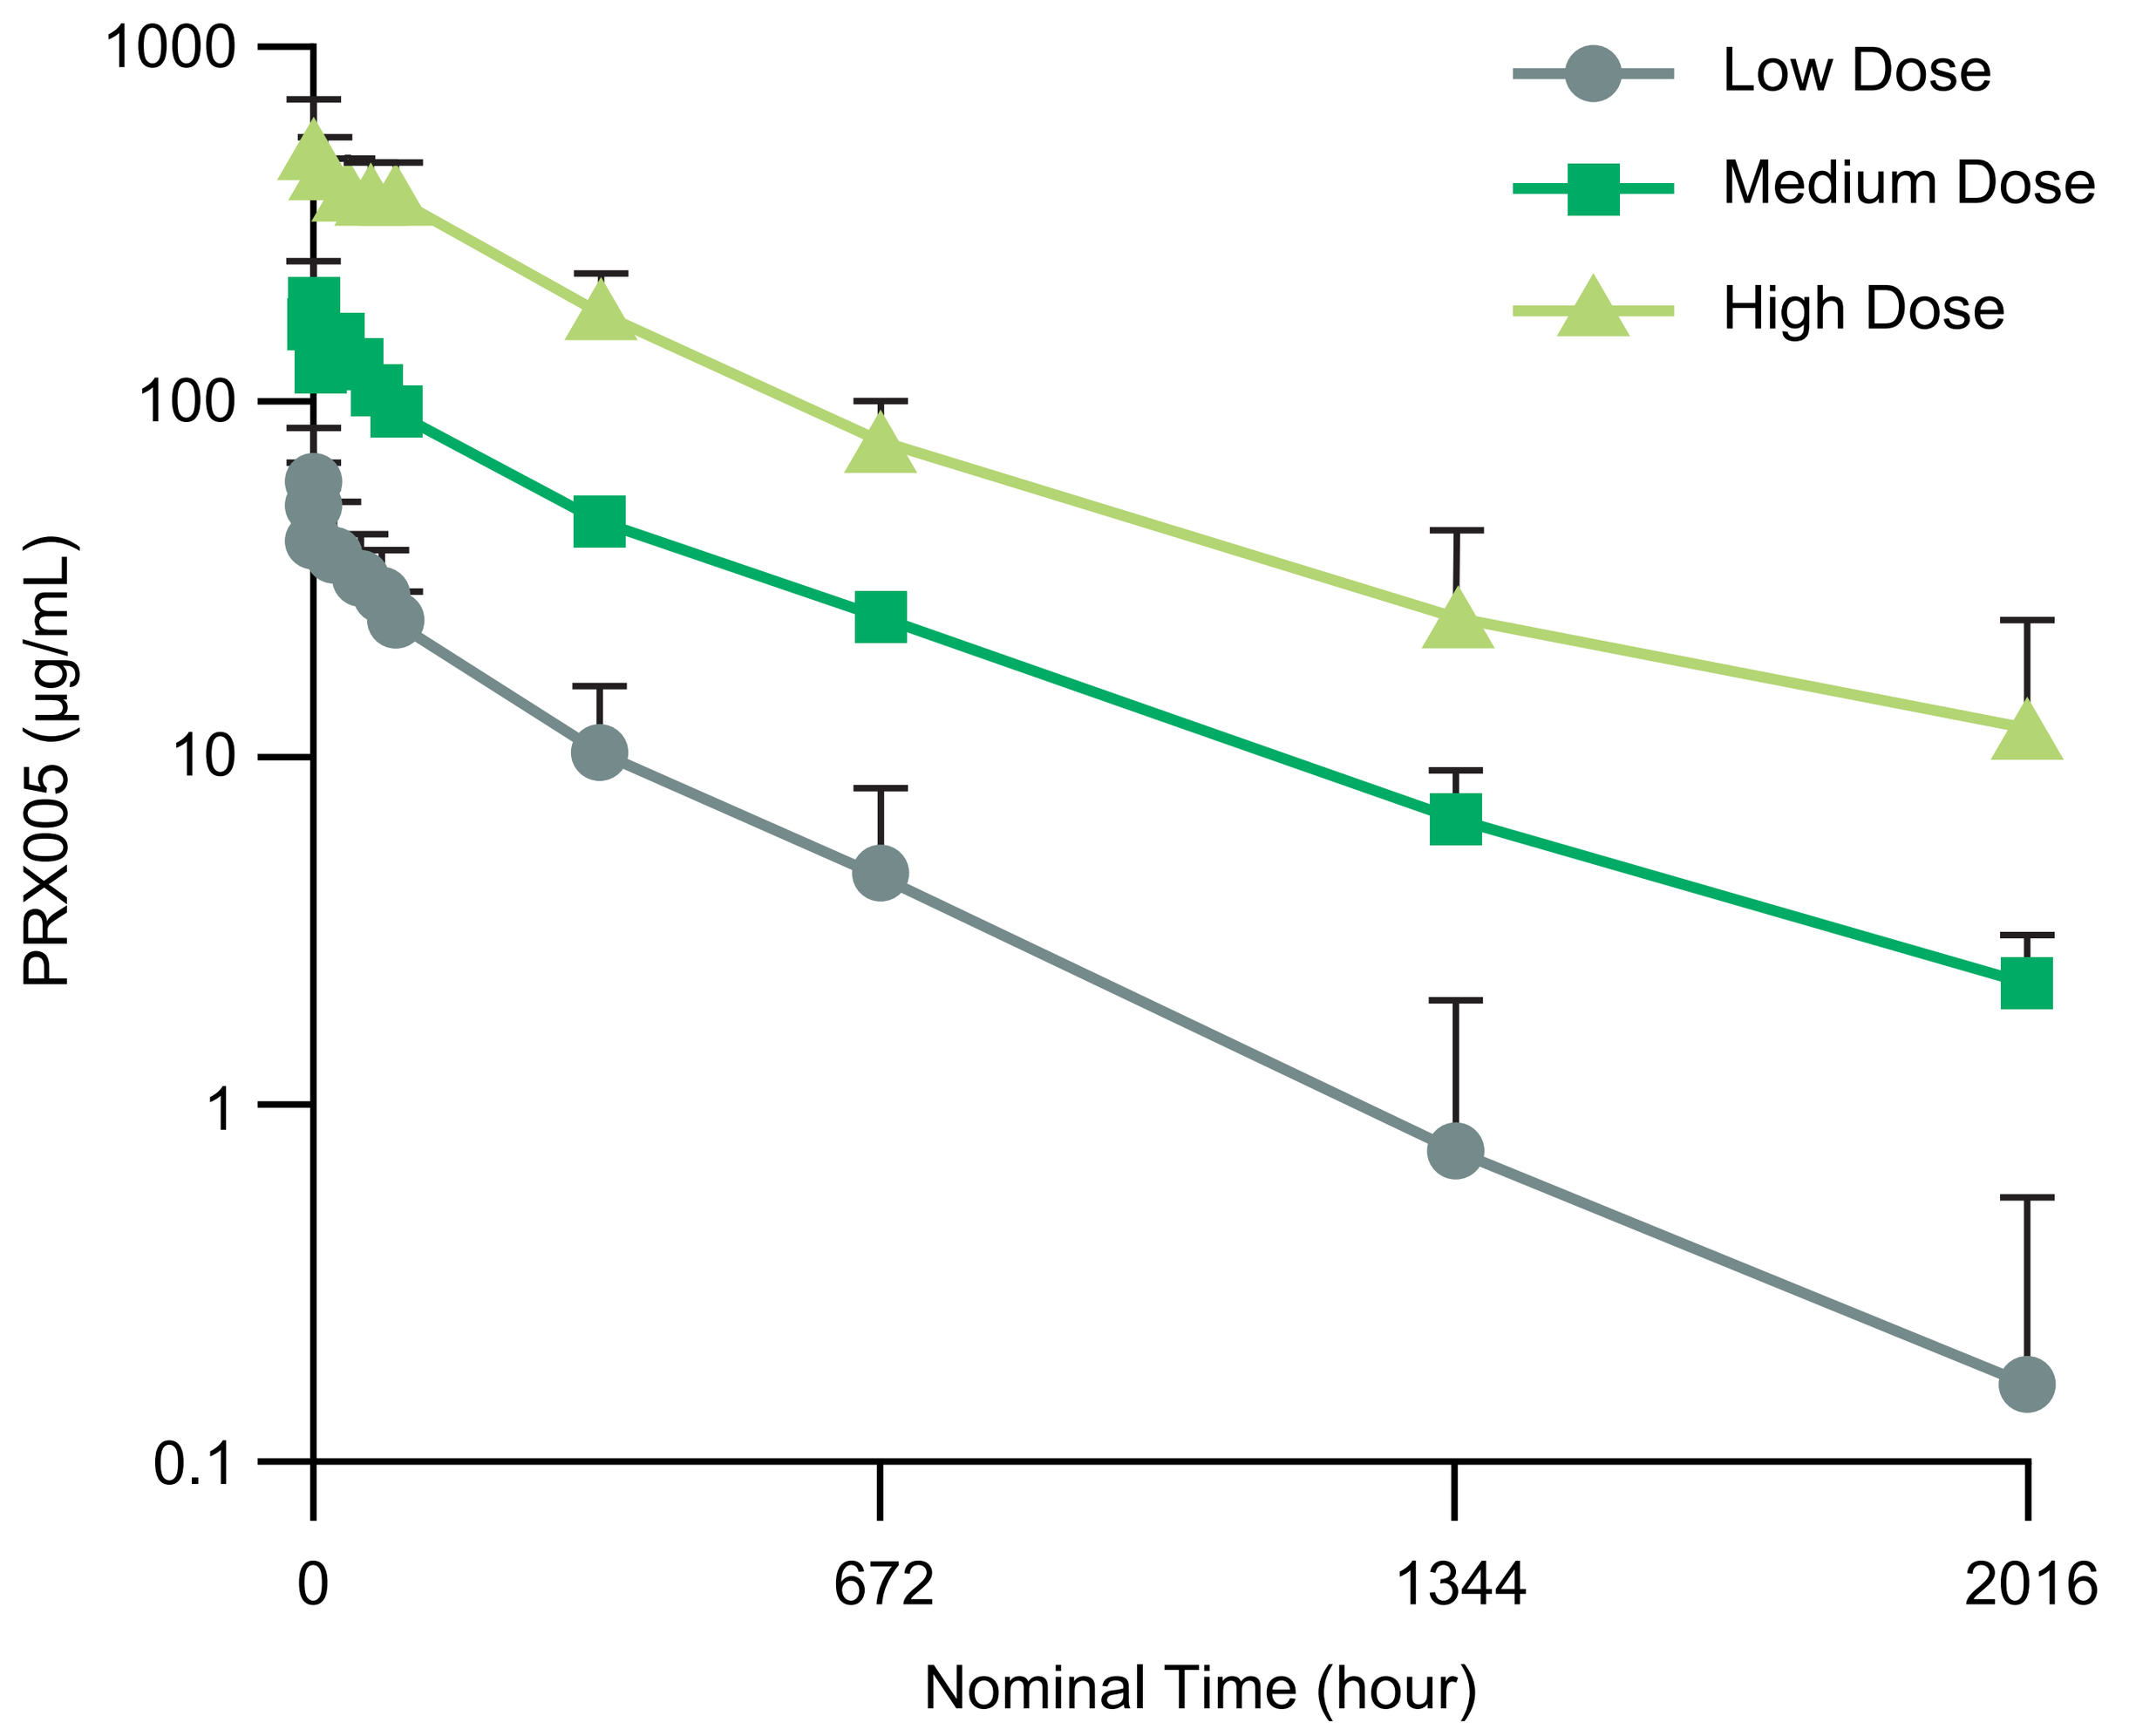
Low Dose
Medium Dose
High Dose
PRX005 (µg/mL)
1000
100
10
1
0.1
0
672
1344
2016
Nominal Time (hour)

Table 3. CSF Drug Levels After a Single Dose

	CSF (ng/mL)	CSF:Plasma (%)
Day 3	298 ± 166[a,*]	0.09 ± 0.081[a,*]
Day 29	166 ± 97[b]	0.20 ± 0.070[b]

Data represent mean ± SD. [a]n=6; [b]n=4; *includes two subjects flagged as out of collection window.

Table 4. Summary of Plasma PK Results: Noncompartmental Analysis

Parameters	Low Dose (n=6)	Medium Dose (n=6)	High Dose (n=6)
T_{max} (hour)	1.86 (0.68)	7.88 (8.99)	2.63 (2.30)
C_{max} (µg/mL)	66.4 (14.3)	202 (42.6)	601 (121)
$AUC_{0-\infty}$ (h*µg/mL)	12,100 (4,750)	53,800 (8,370)	204,000 (34,800)
λ_z (1/hour)	0.0041 (0.0033)	0.00204 (0.00038)	0.00201 (0.00047)
$t_{1/2}$ (day)	10.0 (5.3)	14.5 (2.4)	15.1 (3.7)
CL (L/hour)	0.00029 (0.00013)	0.00019 (0.00003)	0.00015 (0.00003)
Vz (L)	0.0827 (0.0231)	0.0936 (0.0097)	0.0767 (0.0110)

Data represent mean ± SD. Samples for one subject in the low-dose group were not available.

RESULTS (CONTINUED)

Safety Results

- Two of 6 placebo-treated subjects (33.3%) and 10 of 19 PRX005-treated subjects (52.6%) experienced at least one TEAE.
- The majority of TEAEs were not considered related to the study drug; 1 of 6 (16.7%) subjects in the placebo group and 2 of 19 (10.5%) subjects in the PRX005 group had TEAEs that were considered treatment related (**Table 5**).
- No serious adverse events were observed following a single dose of PRX005.

Table 5. Treatment-Related TEAEs by Preferred Term in the Safety Population

	Number of Subjects Reporting (%), Number of Events				
Preferred Term	Placebo (n=6)	PRX005 Low Dose (n=7)	PRX005 Medium Dose (n=6)	PRX005 High Dose (n=6)	PRX005 Combined (n=19)
All treatment-related TEAEs	1 (16.7), 1	2 (28.6), 2	0, 0	0, 0	2 (10.5), 2
Asthenia	0, 0	1 (14.3), 1	0, 0	0, 0	1 (5.3), 1
Headache	0, 0	1 (14.3), 1	0, 0	0, 0	1 (5.3), 1
Orthostatic hypotension	1 (16.7), 1	0, 0	0, 0	0, 0	0, 0

CONCLUSIONS

- We present results of the SAD portion of a first-in-human Phase 1 clinical trial of PRX005, a humanized IgG1 kappa monoclonal tri-epitopic antibody, that binds to the MTBR of tau and was demonstrated preclinically to block cell-to-cell transmission of tau and to reduce intraneuronal tau pathology.
- In this study, PRX005 was generally safe and well tolerated, showed dose-proportional exposure, no treatment-emergent ADAs, and reached sufficient CSF concentrations to predict pharmacological targeting of MTBR tau in the CNS.
- Our results support the further clinical development of PRX005 in an ongoing, multiple ascending dose study in patients with AD.

REFERENCES

1. Kosik KS, et al. *Proc Natl Acad Sci U S A*. 1986;83(11):4044–4048. 2. Vogel JW, et al. *Nat Med*. 2021;27(5):871–881. 3. Nelson PT, et al. J *Neuropathol Exp Neurol*. 2012;71(5):362–381. 4. Dolan P, et al. Oral presentation at 2021 ADPD Conference. 5. Horie K, et al. *Brain*. 2021 Mar 3;144:515–527. 6. Friedhoff P, et al. *Proc Natl Acad Sci U S A*. 1998;95:15712–15717. 7. von Bergen M, et al. *Biochim Biophys Acta*. 2005;1739:158–166.

AUTHOR DISCLOSURES

All authors are employees of Prothena Biosciences Inc and shareholders of Prothena Corporation plc.

ACKNOWLEDGMENTS

This study was sponsored by Prothena Biosciences Limited, Dublin, Ireland, a member of the Prothena Corporation plc group. Editorial support was provided by AMICULUM USA, Princeton, NJ and funded by Prothena Biosciences Inc. We thank our collaborators at Bristol Myers Squibb.

Presented at the Alzheimer's Association International Conference® (AAIC®); July 16–20, 2023; Amsterdam, Netherlands, and Online.

Supported by Prothena Biosciences Limited, Dublin, Ireland.