346

Health-Related Quality of Life (HRQoL) in Patients With Mayo Stage IV Light Chain (AL) Amyloidosis Treated With Birtamimab Plus Standard of Care (SoC): Results From the VITAL Trial

Anita D'Souza,[1] Vaishali Sanchorawala,[2] Efstathios Kastritis,[3] Ashutosh Wechalekar,[4] Stefan Schönland,[5] Raymond Comenzo,[6] Katherine Ingrid Sprinz,[7] Christie Nie,[7] Tim Lin,[7] Morie Gertz[8]

[1]Medical College of Wisconsin, Milwaukee, WI, USA; [2]Boston Medical Center and Boston University Chobanian & Avedisian School of Medicine, Boston, MA, USA; [3]National and Kapodistrian University of Athens, Athens, Greece; [4]University College London, London, UK; [5]Universitätsklinikum Heidelberg, Heidelberg, Germany; [6]Tufts Medical Center, Boston, MA, USA; [7]Prothena Biosciences Inc, Brisbane, CA, USA; [8]Mayo Clinic, Rochester, MN, USA

BACKGROUND

- Systemic AL amyloidosis is a rare, progressive, and typically fatal disease caused by soluble, toxic amyloidogenic light chain aggregates and insoluble light chain amyloid that deposits in vital organs leading to organ dysfunction and failure[1-3]
- Patients with AL amyloidosis have substantially reduced HRQoL compared with other chronic diseases (e.g., chronic lung disease, rheumatoid arthritis)[4,5]
 - Additionally, patients with AL amyloidosis and cardiac involvement have a higher symptom burden and poorer physical function[4,6]
- Birtamimab is an investigational humanized anti-amyloid monoclonal antibody that neutralizes soluble toxic light chain aggregates and clears amyloid deposits from organs[7,8]
 - This contrasts with current SoC, which targets the plasma cell clone but does not address existing soluble, toxic light chain aggregates and insoluble amyloid deposits[3,9]
- The Phase 3 VITAL clinical trial evaluated the efficacy and safety of birtamimab plus SoC versus placebo plus SoC in newly diagnosed, treatment-naïve patients with AL amyloidosis and cardiac involvement[10]
 - VITAL was terminated early after the independent data monitoring committee conducted a futility analysis
 - There was a numerical trend favoring birtamimab in the primary endpoint of time to all-cause mortality (ACM) or cardiac hospitalization for the intention-to-treat population, which was hypothesized to be driven by a treatment effect in the patients with the most significant cardiac involvement (Mayo Stage IV)
 - Post hoc analyses in patients with Mayo Stage IV AL amyloidosis indicated that time to ACM at month 9 improved with birtamimab plus SoC compared with placebo plus SoC (hazard ratio 0.413; 95% confidence interval [CI], 0.191-0.895; *P*=0.021)[10]
 - Additionally, birtamimab plus SoC led to significantly less decline in 36-Item Short Form survey version 2 (SF-36v2) physical component summary (PCS) score versus placebo plus SoC in patients with Mayo Stage IV disease at 9 months, a key secondary endpoint of VITAL[10]
- Here, we assessed longitudinal HRQoL changes across all SF-36v2 domains in patients with Mayo Stage IV AL amyloidosis in VITAL

METHODS

- VITAL was a Phase 3, double-blind, placebo-controlled clinical trial (NCT02312206) in which newly diagnosed treatment-naïve patients with AL amyloidosis received birtamimab plus SoC or placebo plus SoC, as previously described[10]
- Patients in VITAL completed the SF-36v2 at baseline and months 3, 6, and 9
 - Lower SF-36v2 scores indicate worse HRQoL
- All SF-36v2 domains for Mayo Stage IV patients are reported
 - A restricted maximum likelihood-based mixed model for repeated measures was used to estimate least squares mean (LSM), standard error (SE), and 95% CI for each treatment group and LSM difference between groups
 - Fixed effects were included for randomization strata (Renal Stage I vs II/III; 6-minute walk test [6MWT] distance <300 vs ≥300 m), treatment group, categorical time point, and the treatment group x time point interaction, with baseline value included as covariate

RESULTS

Baseline Demographics and Characteristics

- Baseline demographics and characteristics were similar between the birtamimab and placebo arms of Mayo Stage IV patients (**Table 1**)
- Baseline values were also similar between treatment arms for all eight SF-36v2 domains and component summary scores in Mayo Stage IV patients (**Table 2**)
 - Patients with Mayo Stage IV AL amyloidosis tended to have numerically lower average scores than previously reported for the entire AL amyloidosis population,[4–6] as would be expected in patients with greater disease burden[6]

RESULTS

Change in SF-36v2 Domains Over Time

- The LSM change in SF-36v2 domains from baseline to months 3, 6, and 9 for patients with Mayo Stage IV AL amyloidosis is shown in **Figure 1**
 - Significantly less decline was observed at month 6 and 9 in the birtamimab versus placebo arm in social functioning, and at month 9 in role physical, bodily pain, and PCS score
- As shown in **Figure 2**, the LSM differences between treatment arms at month 9 demonstrated significantly less decline in the birtamimab arm than in the placebo arm for role physical, bodily pain, social functioning, and PCS score in patients with Mayo Stage IV AL amyloidosis (PCS reported in Gertz et al. 2023)[10]

Table 1: Baseline demographics and characteristics in patients with Mayo Stage IV AL amyloidosis

	Mayo Stage IV Patients (n=77)	
	Birtamimab + SoC (n=38)	Placebo + SoC (n=39)
Age (years), median (Q1, Q3)	63.6 (55.7, 69.8)	63.7 (57.0, 68.4)
Gender, n (%)		
Male	25 (66)	28 (72)
Female	13 (34)	11 (28)
Ethnicity, n (%)		
Not Hispanic or Latino	34 (90)	36 (92)
Hispanic or Latino	0	0
Not provided or unknown	4 (11)	3 (8)
Race, n (%)		
White	36 (95)	36 (92)
Black or African American	2 (5)	2 (5)
Asian	0	0
Other	0	1 (3)
Age at AL amyloidosis diagnosis (years), median (Q1, Q3)	63.5 (55.6, 69.7)	63.8 (56.8, 68.5)
Duration since AL amyloidosis diagnosis (months), median (Q1, Q3)	1.15 (0.69, 1.58)	1.45 (0.89, 1.81)
Number of derived involved organs at baseline, median (Q1, Q3)	1.5 (1.0, 2.0)	2.0 (1.0, 2.0)
Baseline NT-proBNP ≥1800 pg/mL, n (%)	38 (100)	39 (100)
Baseline NT-proBNP (pg/mL), median (Q1, Q3)	5141.3 (3228.0, 5939.4)	5415.0 (4054.0, 8073.0)
Baseline troponin T (ng/mL),* median (Q1, Q3)	0.05 (0.04, 0.09)	0.09 (0.06, 0.13)
Baseline FLC ratio, median (Q1, Q3)	0.05 (0.02, 0.08)	0.05 (0.03, 11.14)
Baseline dFLC (mg/dL),† median (Q1, Q3)	44.44 (25.13, 56.17)	57.42 (35.52, 106.28)
Mayo Stage, n (%)		
I	NA	NA
II	NA	NA
III	NA	NA
IV	38 (100)	39 (100)
Renal Stage, n (%)		
I	28 (74)	29 (74)
II	9 (24)	9 (23)
III	1 (3)	1 (3)
Baseline 6MWT distance, n (%)		
<300 m	13 (34)	16 (41)
≥300 m	25 (66)	23 (59)

Previously published in Gertz et al. 2023.[10]
*Mayo stage criteria for troponin T levels were modified from a value of 0.025 ng/mL, cited in Kumar et al. 2012,[11] to 0.03 ng/mL, the lowest validated determination for the commercially available test; †Baseline dFLC is calculated only for patients with an abnormal baseline FLC ratio (κ:λ <0.26 or >1.65).
6MWT, 6-minute walk test; AL, light chain; dFLC, difference between involved and uninvolved free light chains; FLC, free light chain; NA, not applicable; NT-proBNP, N-terminal pro-brain natriuretic peptide; Q1, first quartile; Q3, third quartile; SoC, standard of care.

Table 2: Baseline SF-36v2 scores in patients with Mayo Stage IV AL amyloidosis

	Mayo Stage IV Patients (n=77)	
	Birtamimab + SoC (n=38)	Placebo + SoC (n=39)
Baseline SF-36v2 score, mean (SD)		
Physical functioning	35.7 (24.4)	32.5 (22.0)
Role physical	31.6 (23.4)	32.2 (32.6)
Bodily pain	60.1 (26.3)	66.5 (27.4)
General health	41.6 (18.0)	43.2 (17.1)
Vitality	36.4 (24.5)	34.2 (20.6)
Social functioning	51.0 (31.2)	63.2 (27.9)
Role emotional	76.1 (26.1)	78.1 (28.8)
Mental health	68.7 (14.7)	70.5 (19.6)
Physical component summary	33.6 (8.8)	33.8 (10.0)
Mental component summary	48.2 (9.5)	50.1 (11.3)

Baseline is the last non-missing assessment prior to the first infusion of study drug.
AL, light chain; SD, standard deviation; SF-36v2, 36-Item Short Form survey version 2; SoC, standard of care.

Figure 1: LSM change in SF-36v2 scores from baseline to months 3, 6, and 9 in patients with Mayo Stage IV AL amyloidosis

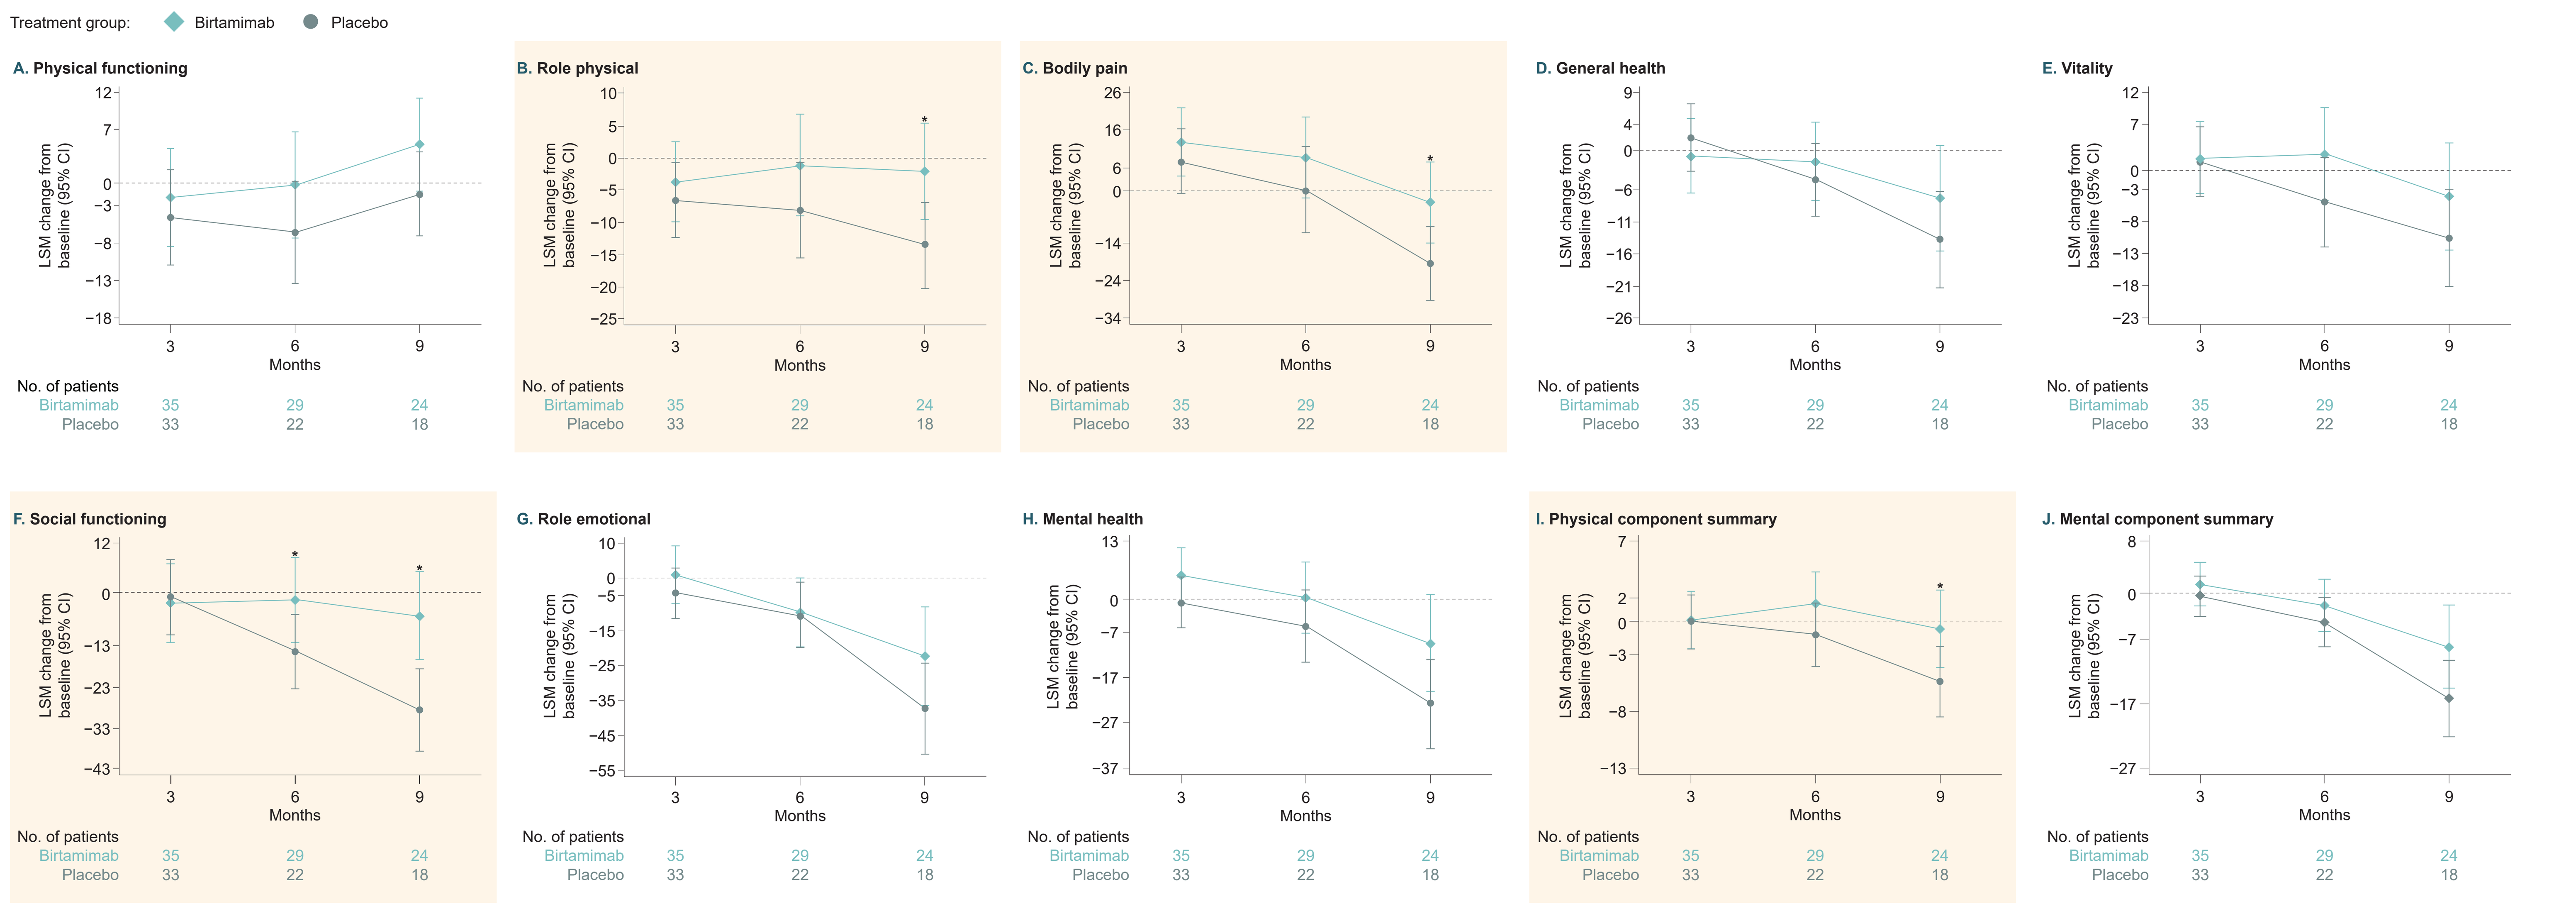
Treatment group: Birtamimab Placebo
A. Physical functioning
B. Role physical
C. Bodily pain
D. General health
E. Vitality
F. Social functioning
G. Role emotional
H. Mental health
I. Physical component summary
J. Mental component summary
LSM change from baseline (95% CI)
Months
No. of patients
Birtamimab 35 29 24
Placebo 33 22 18

Highlighted figures demonstrate significantly less decline in the birtamimab arm versus the placebo arm.
*$P<0.05$.
AL, light chain; CI, confidence interval; LSM, least-squares mean; SF-36v2, 36-Item Short Form survey, version 2.

Figure 2: Forest plot of change from baseline to month 9 in SF-36v2 domains in patients with Mayo Stage IV AL amyloidosis

SF-36v2 domain	LSM difference (95% CI)	*P* value
Physical functioning	6.6 (−1.40, 14.59)	0.106
Role physical	11.5 (1.56, 21.34)	0.023
Bodily pain	16.3 (2.02, 30.66)	0.025
General health perceptions	6.4 (−4.48, 17.29)	0.248
Vitality	6.4 (−4.58, 17.45)	0.252
Social functioning	22.8 (8.26, 37.32)	0.002
Role emotional	15.0 (−4.14, 34.22)	0.124
Mental health	13.4 (−1.12, 27.82)	0.070
Physical component summary	4.7 (0.08, 9.22)	0.046
Mental component summary	8.0 (−0.79, 16.81)	0.074

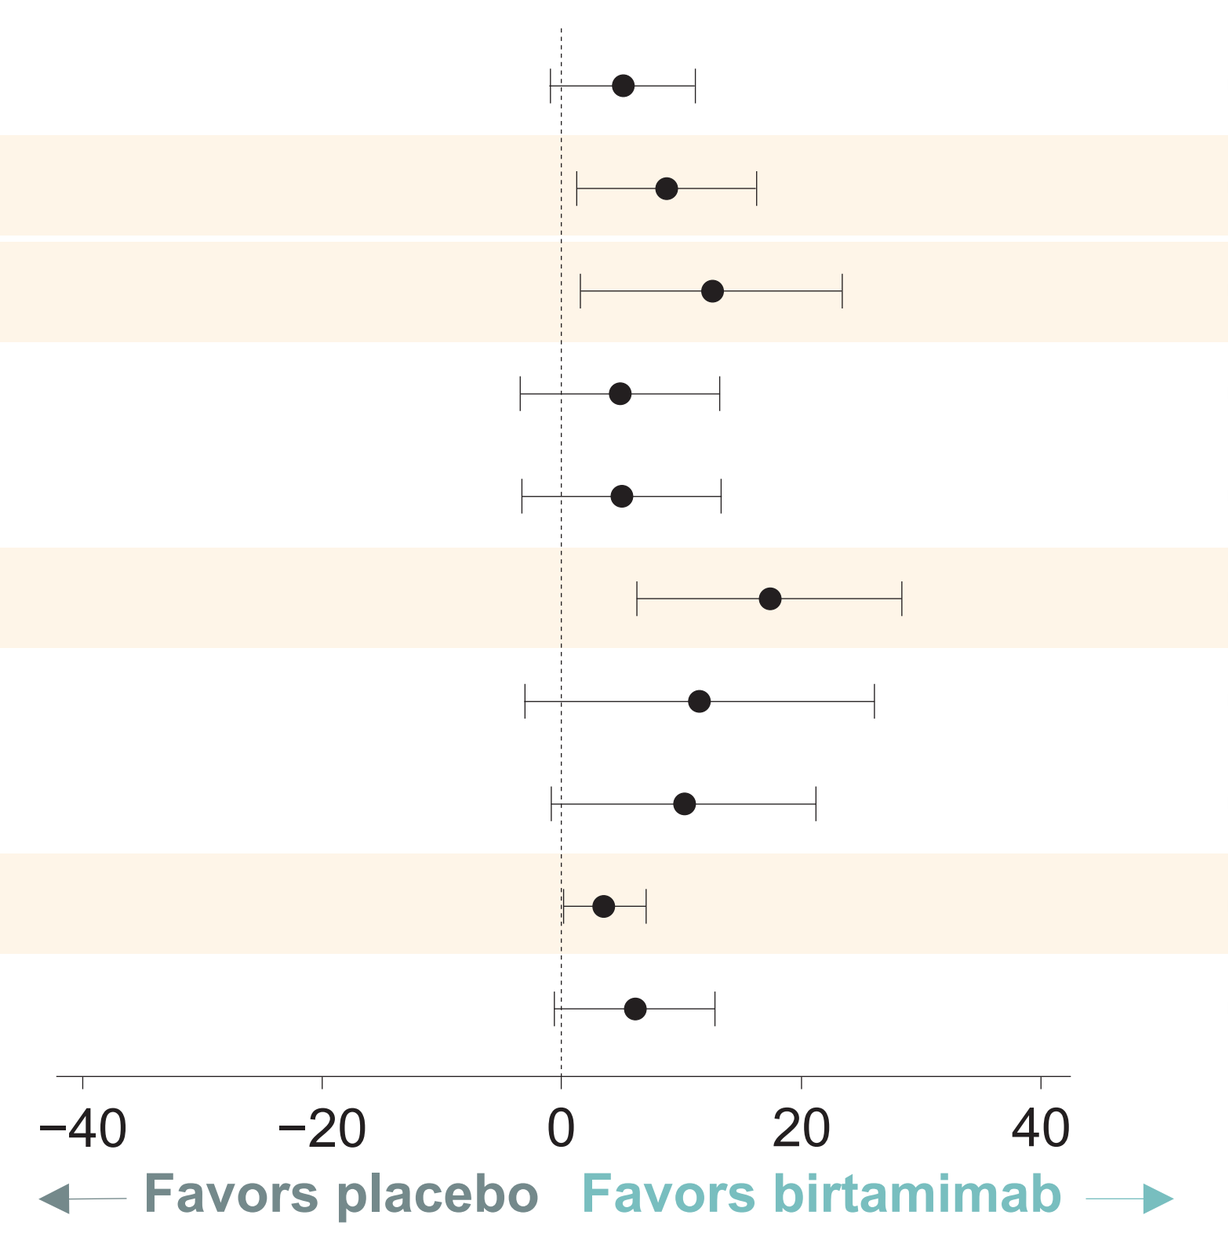
−40 −20 0 20 40
Favors placebo
Favors birtamimab

Birtamimab-treated patients at month 9, n=24; placebo patients at month 9, n=18.
AL, light chain; CI, confidence interval; LSM, least-squares mean; SF-36v2, 36-Item Short Form survey, version 2.

CONCLUSIONS

- In a post hoc analysis of the VITAL clinical trial, birtamimab plus SoC showed a survival benefit in patients with Mayo Stage IV AL amyloidosis, a subgroup of patients with a higher symptom burden and poorer physical functioning[4,6,10]
 - The survival benefit of birtamimab plus SoC in patients with Mayo Stage IV AL amyloidosis is being further explored in the AFFIRM-AL clinical trial (NCT04973137) under a Special Protocol Assessment (SPA) agreement with the US Food and Drug Administration (primary endpoint ACM at a significance level of 0.10)[10,12]
- In the present analysis, all SF-36v2 domain scores at month 9 numerically favored birtamimab plus SoC over placebo plus SoC in patients with Mayo Stage IV AL amyloidosis
 - Additionally, birtamimab plus SoC was associated with significantly less decline in HRQoL versus placebo plus SoC in role physical, bodily pain, social functioning, and PCS score
- The ongoing confirmatory Phase 3 AFFIRM-AL clinical trial will collect additional SF-36v2 data that may enable better understanding of the impact of birtamimab on HRQoL in patients with Mayo Stage IV AL amyloidosis[12]

REFERENCES

1. Vaxman I, Gertz M. *Acta Haematol.* 2019;141:93-106.
2. Hwa YL, et al. *J Adv Pract Oncol.* 2019;10:470-481.
3. Muchtar E, et al. *Mayo Clin Proc.* 2021;96:1546-1577.
4. Bayliss M, et al. *Orphanet J Rare Dis.* 2017;12:15.
5. Lousada I, et al. *Orphanet J Rare Dis.* 2020;15:268.
6. D'Souza A, et al. *Eur J Haematol.* 2023;111:536-543.
7. Renz M, et al. *Amyloid.* 2016;23:168-177.
8. Wall JS, et al. *PLoS One.* 2012;7:e52686.
9. Wechalekar AD, et al. *Amyloid.* 2023;30:3-17.
10. Gertz MA, et al. *Blood.* 2023;142:1208-1218.
11. Kumar S, et al. *J Clin Oncol.* 2012;30:989-995.
12. AFFIRM-AL Study. NCT04973137. Accessed April 2024. https://clinicaltrials.gov/study/NCT04973137.

AUTHOR DISCLOSURES

AD: Imbrium, Pfizer, Bristol Myers Squibb: Membership on an entity's Board of Directors or advisory committees; Janssen, Prothena: Consultancy; AbbVie, Sanofi, Takeda, TeneoBio, Caelum, Prothena: Research Funding. **VS:** Janssen, Alexion, Prothena, Celgene, Takeda, AbbVie, Regeneron, Pfizer, AstraZeneca: Consultancy, Honoraria, Membership on an entity's Board of Directors or advisory committees, Research Funding. **EK:** Sanofi: Honoraria; Pfizer, GSK, Janssen: Honoraria, Research Funding. **AW:** Alexion/AstraZeneca, Attralus, GSK, Janssen, Pfizer, Prothena: Consultancy, Honoraria. **SS:** Janssen, Takeda, Pfizer, Prothena: Honoraria; Prothena, Janssen, Sanofi: Research Funding; Janssen, Prothena, Celgene, Binding Site, Jazz: Other: Travel grant. **RC:** Nothing to disclose. **KIS, CN, and TL:** Prothena Biosciences, Inc: Current employment; Prothena Corporation plc: Current equity holder in publicly-traded company. **MG:** Ionis/Akcea, Prothena, Sanofi, Janssen, Aptitude Healthgrants, Ashfield, Physicians Education Resource, Research to Practice, Johnson & Johnson, Celgene: Consultancy; AbbVie: Other: Data Safety Monitoring board; i3HEalth: Other: For development of educational material; Juno Pharmaceutics, Sorrento Therapeutics: Other: Meetings.

ACKNOWLEDGEMENTS

The authors thank the patients who participated in the VITAL clinical trial, the study sites and investigators, and the Data Monitoring Committee.
This poster was sponsored by Prothena Biosciences Ltd, Dublin, Ireland, a member of the Prothena Corporation plc group.
Medical writing support was provided by Teal M. Brechtel, PhD of AMICULUM USA, and funded by Prothena Biosciences Ltd.

Presented at XIX International Symposium on Amyloidosis, May 26–30, 2024, Rochester, Minnesota, USA

Presenter: Dr. Anita D'Souza (anitadsouza@mcw.edu)